Nintedanib in IPF: post hoc analysis of the Italian FIBRONET observational study

Venerino Poletti,[1,2] Carlo Albera,[3] Sergio Harari,[4] Alberto Pesci,[5] Alessandra Ori,[6] Giovanna Crespi,[7] Benedetta Campolo,[7] Carlo Vancheri[8]

1. Department of Diseases of the Thorax, Ospedale GB Morgagni, Forlì, Italy; 2. Department of Respiratory Diseases & Allergy, Aarhus University Hospital, Aarhus, Denmark; 3. S.C.U. Pneumologia, A.O.U. Città Della Scienza e della Salulte (Molinette), University of Torino, Italy; 4. Department of Clinical Sciences and Community Health, University of Milan, Italy; Department of Medicine, Ospedale San Giuseppe MultiMedica IRCCS, Milan, Italy; 5. Ospedale San Gerardo, ASST Monza, Monza, Italy; 6. MediNeos Observational Research, Modena, Italy; 7. Boehringer Ingelheim, Milano, Italy; 8. Regional Referral Centre for Rare Lung Diseases – A.O.U. Policlinico-Vittorio Emanuele, University of Catania, Catania, Italy

BACKGROUND

FIBRONET (NCT02803580)

- Real-world, observational study of patients with IPF in Italy

N=209

Mean (±SD) FVC% pred:
- **80.0%** (±19.2) at baseline
- **82.2%** (±20.9) at 12 months

12 months

47.4% of patients:
- No decline in FVC% pred during study

20 centres

Antifibrotic therapy (nintedanib or pirfenidone)
- Patients receiving at 12 months: **83.9%**
- Mean time from diagnosis to treatment: **6.4 weeks**

AIM: To conduct a post hoc analysis of patients who received nintedanib during the FIBRONET study

INCLUSION CRITERIA

Diagnosed with IPF ≤3 months

Treated with nintedanib for ≥7 months

Available FVC% pred values at baseline and 12 months:[1]
- Intermediate evaluations at 3, 6 and 9 months

RESULTS

1. Baseline characteristics

N=52

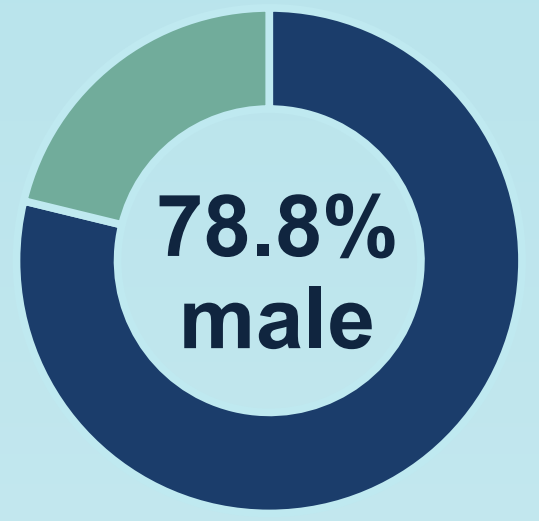
78.8%
male

≥1 comorbidity:
- **84.6%** of patients

- Age: **70.2** (±7.1) **years**
- Time from first IPF diagnosis to enrolment: **0.9** (±1.1) **months**
- Duration of nintedanib treatment: **11.6** (±1.6) **months**

(All values mean [±SD])

Mean (±SD) FVC% pred at baseline: **78.7%** (±15.0)

42.3% of patients had FVC ≥80% pred

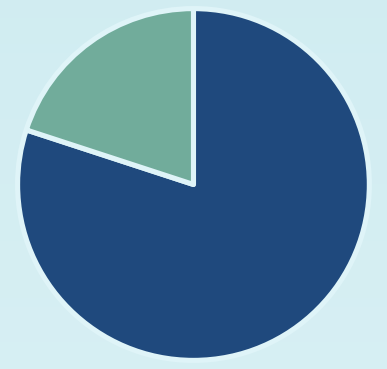

Most patients received a full dose of nintedanib during the 12 months:
- 76.9%: 150 mg BID nintedanib
- 19.2%: 100 mg BID (reduced dose to manage AEs)[2]

2. Change in lung function

Mean (±SD) FVC% pred at 12 months:
- **79.8%** (±15.5) (n=52)

Proportion of patients with:
- ≥5% decline in FVC% pred: **25.0%**
- ≥10% decline: **15.3%**
- <5% decline or increase: **75.0%**

In the 10 patients who had a **dose reduction** (150 mg → 100 mg BID), mean (±SD) FVC% pred:
- Baseline: **77.7%** (±20.0)
- 12 months: **81.0%** (±16.7)

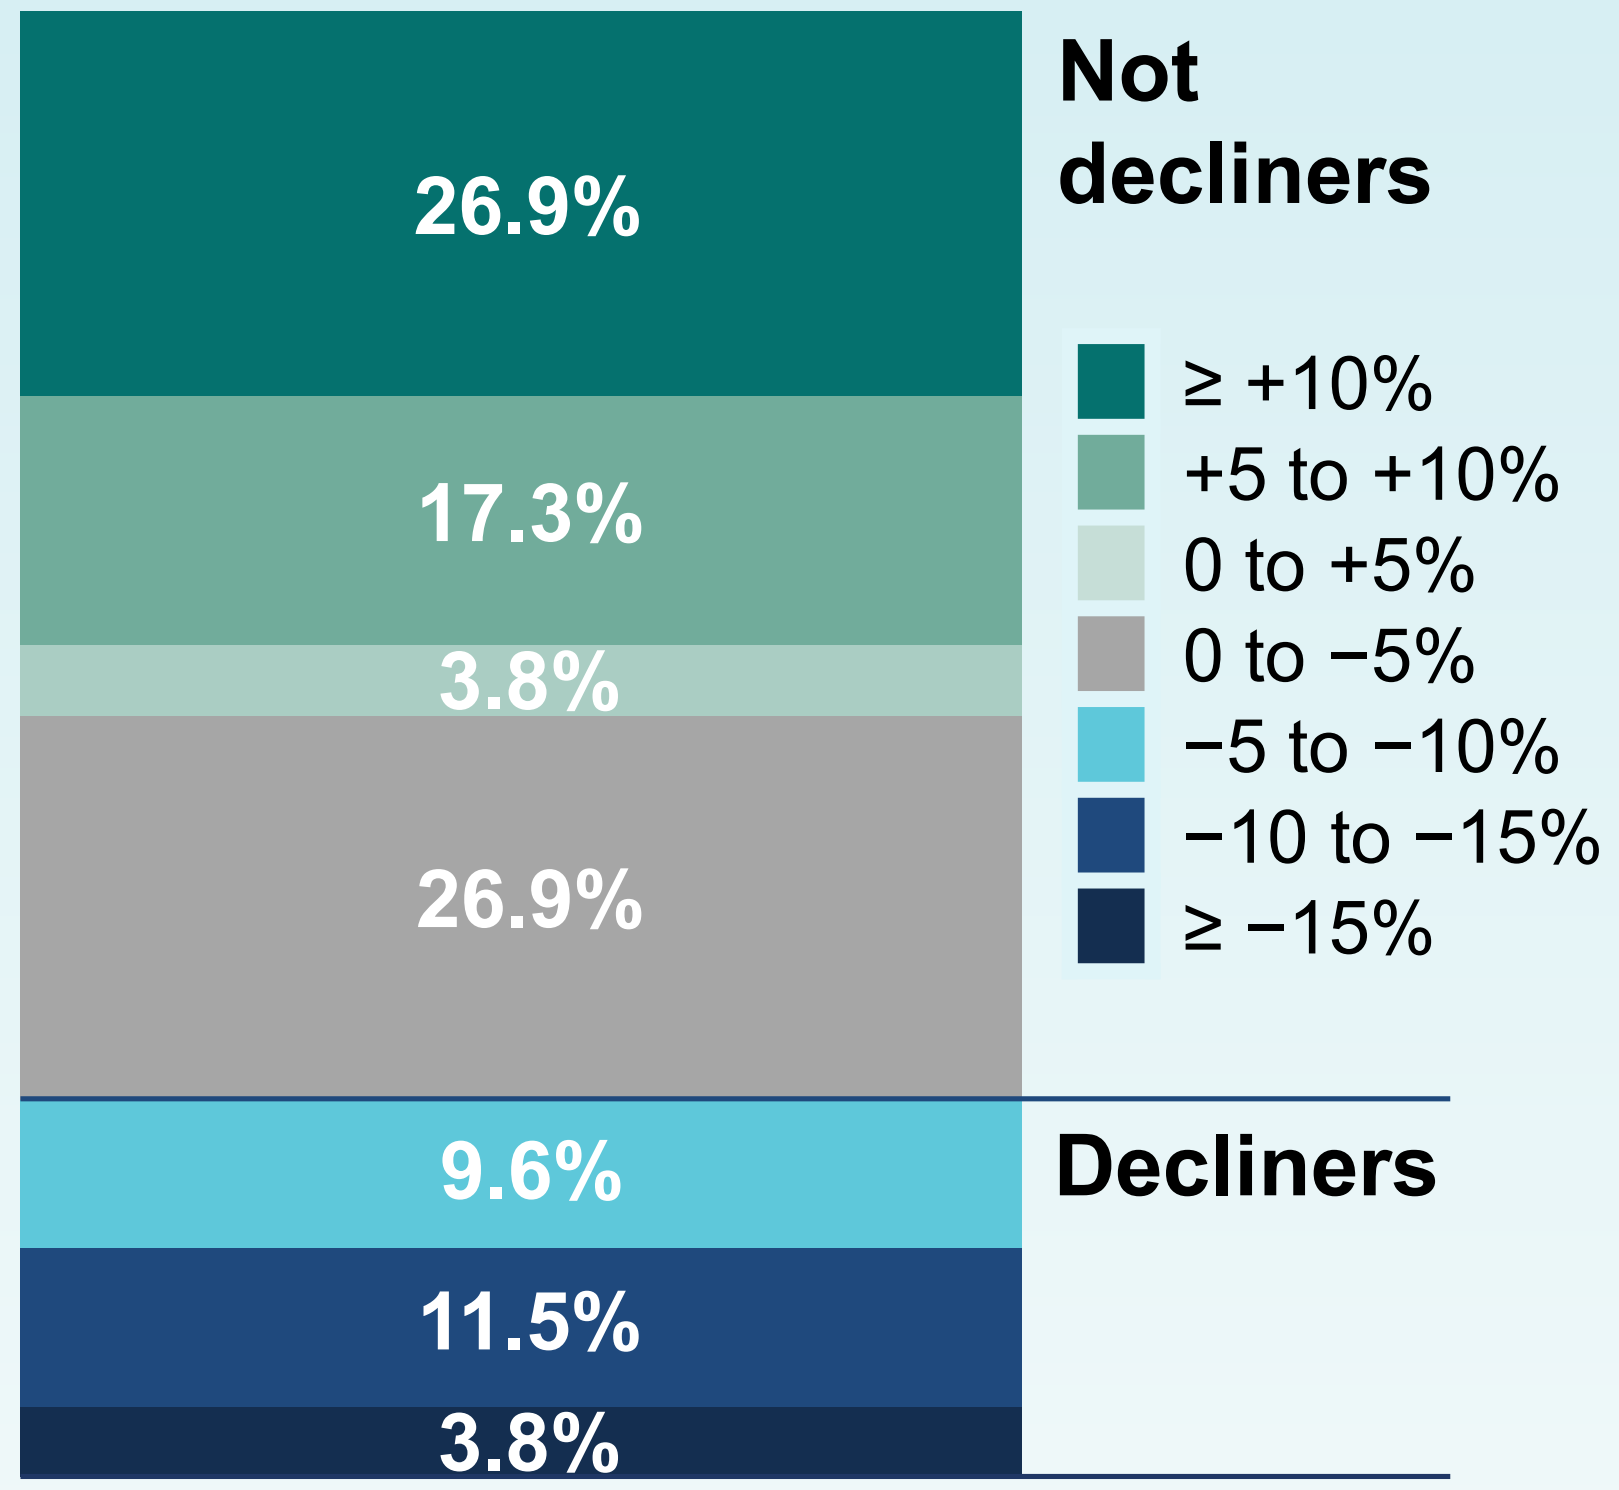
26.9%
17.3%
3.8%
26.9%
9.6%
11.5%
3.8%
Not decliners
Decliners
≥ +10%
+5 to +10%
0 to +5%
0 to −5%
−5 to −10%
−10 to −15%
≥ −15%

Proportion of patients with categorical changes in FVC% pred during 12 months of observation

3. Anxiety/depression, coughing, acute exacerbations, AEs

Mean (±SD) total HADS score

Baseline: **11.5** (±6.9) (n=44)

6 months: **12.3** (±6.9) (n=36)

12 months: **10.8** (±8.7) (n=36)

Max HADS score (depression/anxiety combined): 42

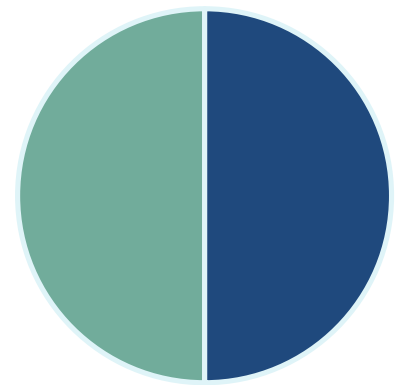

Baseline: **50.0%** of patients had cough (n=26)

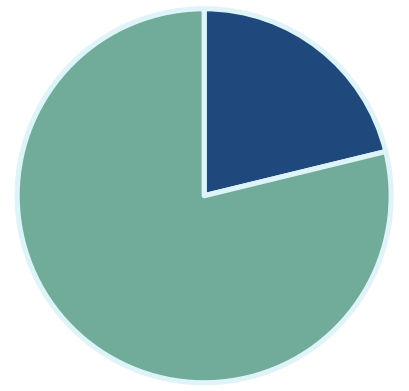

12 months: **21.2%** of patients had cough[a] (n=11)

[a]Majority of patients had coughing symptoms at some, but not all, visits.

Two patients (3.8%) had ≥1 acute exacerbation

Four exacerbations in total; two 'moderate' and two 'severe' (classified according to clinical judgement)

AEs occurring at a frequency of >5% during the observation period, AEs leading to discontinuation of nintedanib, and all SAEs

	No. of patients (%)
Any AE	27 (51.9)
Diarrhoea	18 (34.6)
Lack of appetite	3 (5.8)
Weight loss	3 (5.8)
SAEs	2 (3.8)[3]
AEs leading to discontinuation of nintedanib	2 (3.8)[4]

CONCLUSIONS

- In patients with IPF who received nintedanib for ≥7 months in the FIBRONET study, FVC was stable over 12 months
- There was a low rate of discontinuation, and the safety profile observed was consistent with the known safety profile for nintedanib in IPF

INTERACTIVE

https://www.globalmedcomms.com/respiratory/ERS2020

https://www.globalmedcomms.com/respiratory/ERS2020/Poletti

Footnotes

[1]Missing FVC% pred data at 12 months were imputed using a 'last observation carried forward' approach.
[2]The remaining two patients had a decrease in the frequency of administration (from 150 mg BID to 150 mg QD) and an increase in the dosage of nintedanib (from 100 mg BID to 150 mg BID), respectively.
[3]One patient had angina and a myocardial infarction leading to death; another patient had respiratory insufficiency.
[4]One patient discontinued due to diarrhoea and nausea; another due to lack of appetite and weight loss.

Abbreviations

AE, adverse event; BID, twice daily; FVC, forced vital capacity; HADS, Hospital Anxiety and Depression Scale; IPF, idiopathic pulmonary fibrosis; QD, once daily; SAE, serious adverse event; SD, standard deviation.

Author disclosures

VP has received personal fees from Boehringer Ingelheim. CA has acted as a clinical trial investigator for Boehringer Ingelheim and Roche, participated in scientific advisoryboards for Boehringer Ingelheim, Roche, MSD, Fibrogenand GSK, and received research grants from Boehringer Ingelheim and Roche. SH has acted as a clinical trial investigator, participated in scientific advisory boards, and delivered lectures for Boehringer Ingelheim, Roche and Actelion. He has also received research grants from Boehringer Ingelheim. AP has received personal (speaker/advisory board) fees from Boehringer Ingelheim and Roche, and his research activity is partially supported by project Premia. AO is an employee of Medineos Observational Research. GC and BC are employees of Boehringer Ingelheim (Italy). CV has received research grants and personal fees from Boehringer Ingelheim and Roche.

Acknowledgements

This study was supported by Boehringer Ingelheim International GmbH (BI). The authors meet criteria for authorship as recommended by the International Committee of Medical Journal Editors (ICMJE). The authors did not receive payment for the development of the poster. Writing, editorial support and formatting assistance was provided by Chester Trinick, of MediTech Media, UK, which was contracted and funded by BI. BI was given the opportunity to review the poster for medical and scientific accuracy as well as intellectual property considerations.

Boehringer Ingelheim

Poster presented at the ERS International Virtual Congress, 7–9 September 2020