OP0115

Effect of nintedanib on progression of interstitial lung disease (ILD) in patients with autoimmune disease-related ILDs: further data from the INBUILD® trial

Eric L. Matteson,[1] Clive Kelly,[2] Jörg H.W. Distler,[3] Anna Maria Hoffmann-Vold,[4] James R. Seibold,[5] Shikha Mittoo,[6] Oliver Distler,[7] Paul F. Dellaripa,[8] Alexandra James,[9] Rozsa Schlenker-Herceg,[10] Susanne Stowasser,[9] Manuel Quaresma,[9] Kevin R. Flaherty[11] on behalf of the INBUILD trial investigators

[1]Mayo Clinic College of Medicine and Science, Rochester, MN, USA; [2]Institute of Cellular Medicine, Newcastle University, Newcastle upon Tyne, UK; [3]University of Erlangen-Nuremberg, Erlangen, Germany; [4]Department of Rheumatology, Oslo University Hospital, Oslo, Norway; [5]Scleroderma Research Consultants LLC, Aiken, SC, USA; [6]University Health Network, Toronto, Ontario, Canada; [7]Department of Rheumatology, University Hospital Zurich, Zurich, Switzerland; [8]Division of Rheumatology, Brigham and Women's Hospital, Boston, MA, USA; [9]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany; [10]Boehringer Ingelheim Pharmaceuticals, Inc., Ridgefield, CT, USA; [11]Division of Pulmonary and Critical Care Medicine, University of Michigan, Ann Arbor, MI, USA

Introduction

- Nintedanib is an inhibitor of tyrosine kinases that inhibits processes fundamental to the progression of pulmonary fibrosis.[1]
- The INBUILD trial investigated the efficacy and safety of nintedanib in patients with chronic fibrosing ILDs with a progressive phenotype, excluding idiopathic pulmonary fibrosis (IPF).[2]

Aim

- To assess the effects of nintedanib on the risks of death, acute exacerbation of ILD or death, and disease progression or death in subjects with autoimmune disease-related ILDs in the INBUILD trial.

Methods

Trial design[2]

- Subjects in the INBUILD trial had a physician-diagnosed ILD other than IPF, reticular abnormality with traction bronchiectasis (with or without honeycombing) of >10% extent on HRCT, FVC ≥45% predicted and DLco ≥30%–<80% predicted.
- Subjects met ≥1 of the following criteria for ILD progression in the 24 months before screening, despite management deemed appropriate in clinical practice:

Relative decline in FVC ≥10% predicted	Relative decline in FVC ≥5%–<10% predicted and increased extent of fibrosis on HRCT
Relative decline in FVC ≥5%–<10% predicted and worsened respiratory symptoms	Worsened respiratory symptoms and increased extent of fibrosis on HRCT

- Subjects were randomised to receive nintedanib or placebo. The second database lock took place after all the subjects had completed the post-treatment follow-up visit or entered the open-label extension study.

The INBUILD trial

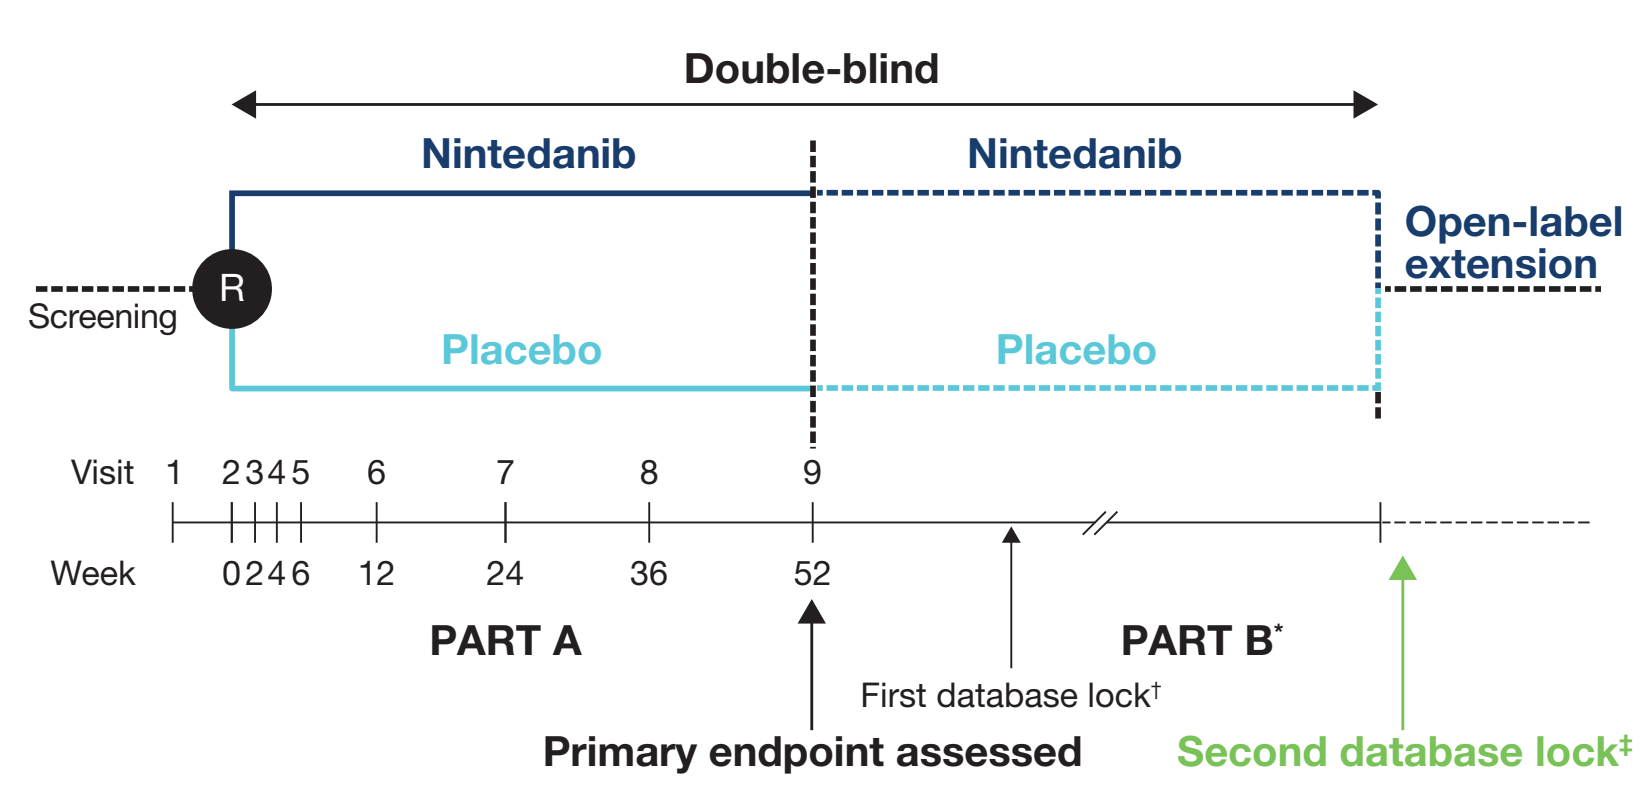
Double-blind
Nintedanib
Nintedanib
Open-label extension
Screening
R
Placebo
Placebo
Visit 1 2345 6 7 8 9
Week 0246 12 24 36 52
PART A
PART B*
First database lock†
Primary endpoint assessed
Second database lock‡

R, Randomisation 1:1, stratified by HRCT pattern (UIP-like fibrotic pattern or other fibrotic patterns).
* Visits occurred every 16 weeks until end of treatment.
† After last subject had completed week 52 visit.
‡ After all patients had completed follow-up visit or entered open-label extension study.

Analyses

- In the subgroup of patients with autoimmune disease-related ILDs, we assessed the following using data up to the second database lock:
 - Time to death
 - Time to first acute exacerbation of ILD or death
 - Time to disease progression (absolute decline in FVC ≥10% predicted) or death
 - Adverse events

Results

Subjects

Subjects with autoimmune disease-related ILDs (n=170)

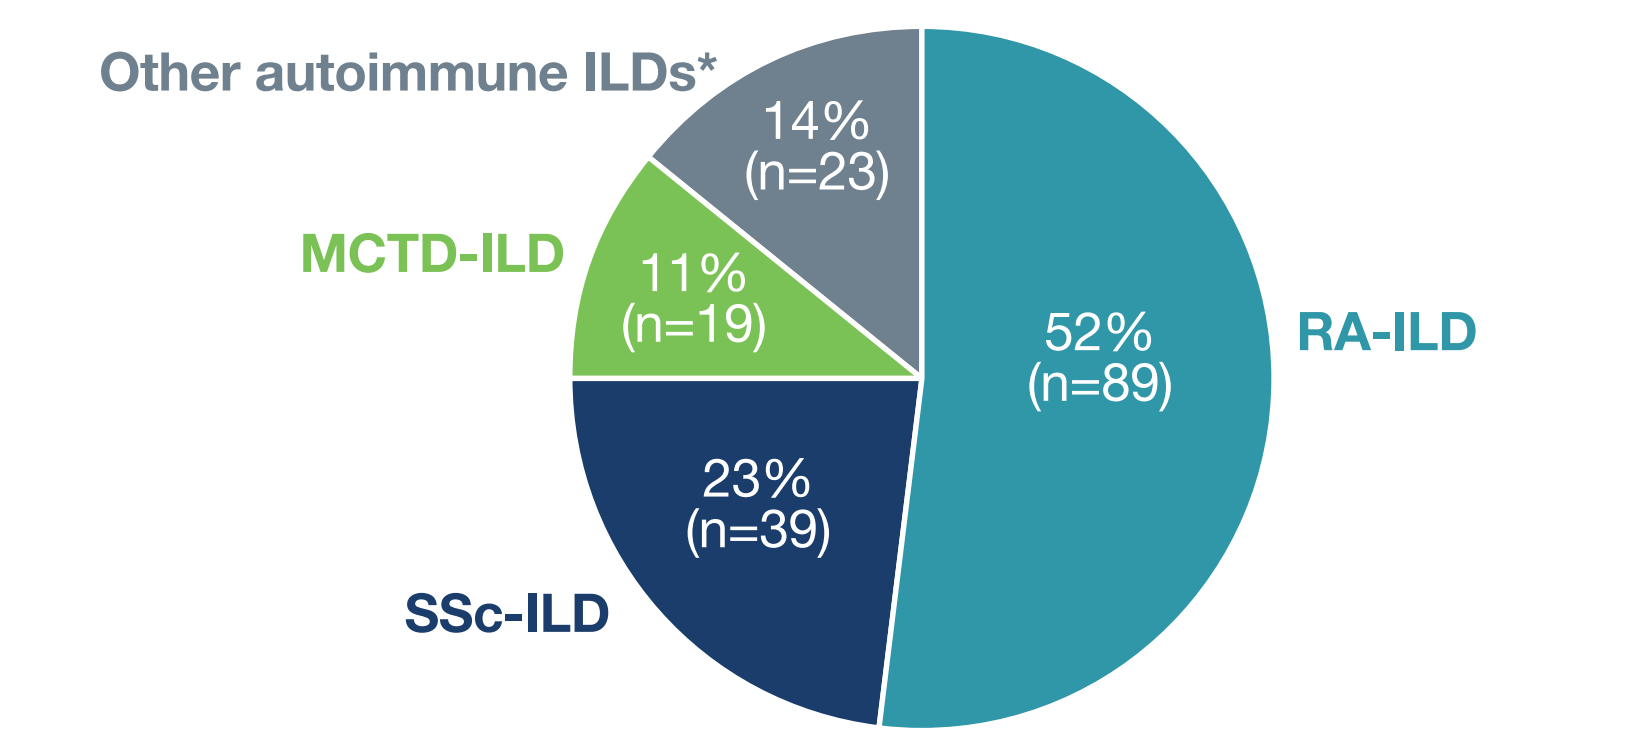
Other autoimmune ILDs*
14% (n=23)
MCTD-ILD
11% (n=19)
52% (n=89)
RA-ILD
23% (n=39)
SSc-ILD

*Subjects with an autoimmune disease noted in the "Other fibrosing ILDs" category of the case report form, including Sjögren's disease-related ILD, IPAF and undifferentiated autoimmune disease-related ILD.
RA, rheumatoid arthritis; SSc systemic sclerosis; MCTD, mixed connective tissue disease; IPAF, interstitial pneumonia with autoimmune features.

Baseline characteristics

53% female	75% UIP-like fibrotic pattern on HRCT
Mean (SD) age 64 (11) years	Mean (SD) FVC 70.9 (14.9) % predicted
50% former or current smokers	Mean (SD) DLco 48.0 (15.1) % predicted

Exposure to trial medication

- Mean (SD) exposure was 15.4 (7.4) months in the nintedanib group and 16.9 (6.1) months in the placebo group.

Time to death

- 8 patients (9.8%) in the nintedanib group and 11 patients (12.5%) in the placebo group died.

Time to death

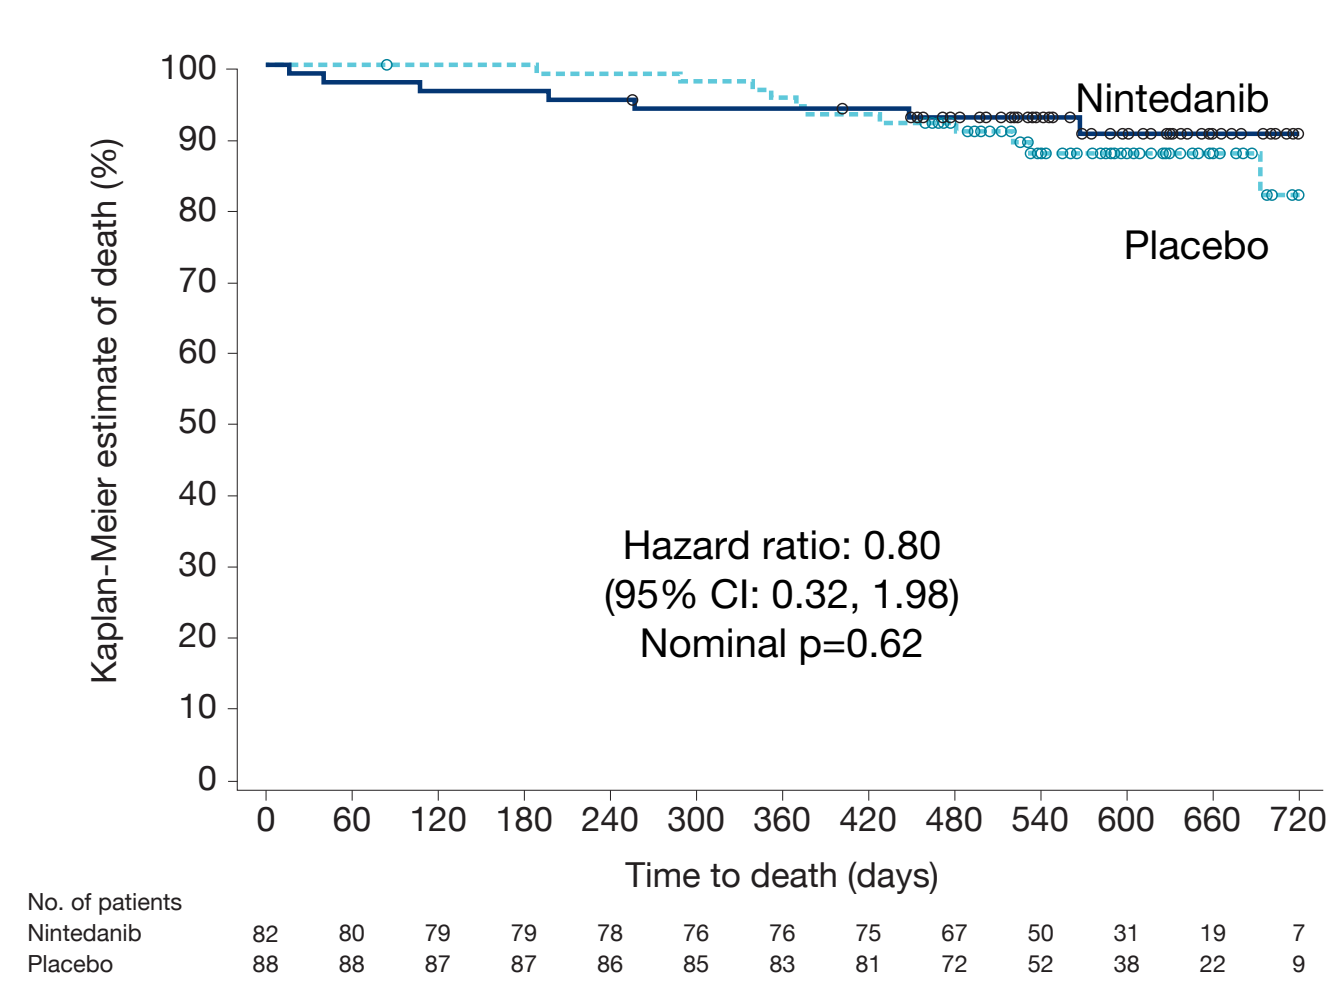
Kaplan-Meier estimate of death (%)
100 90 80 70 60 50 40 30 20 10 0
Nintedanib
Placebo
Hazard ratio: 0.80
(95% CI: 0.32, 1.98)
Nominal p=0.62
0 60 120 180 240 300 360 420 480 540 600 660 720
Time to death (days)
No. of patients
Nintedanib 82 80 79 79 78 76 76 75 67 50 31 19 7
Placebo 88 88 87 87 86 85 83 81 72 52 38 22 9

Time to first acute exacerbation of ILD or death

- 10 patients (12.2%) in the nintedanib group and 18 patients (20.5%) in the placebo group had an acute exacerbation of ILD or died.

Time to first acute exacerbation of ILD or death

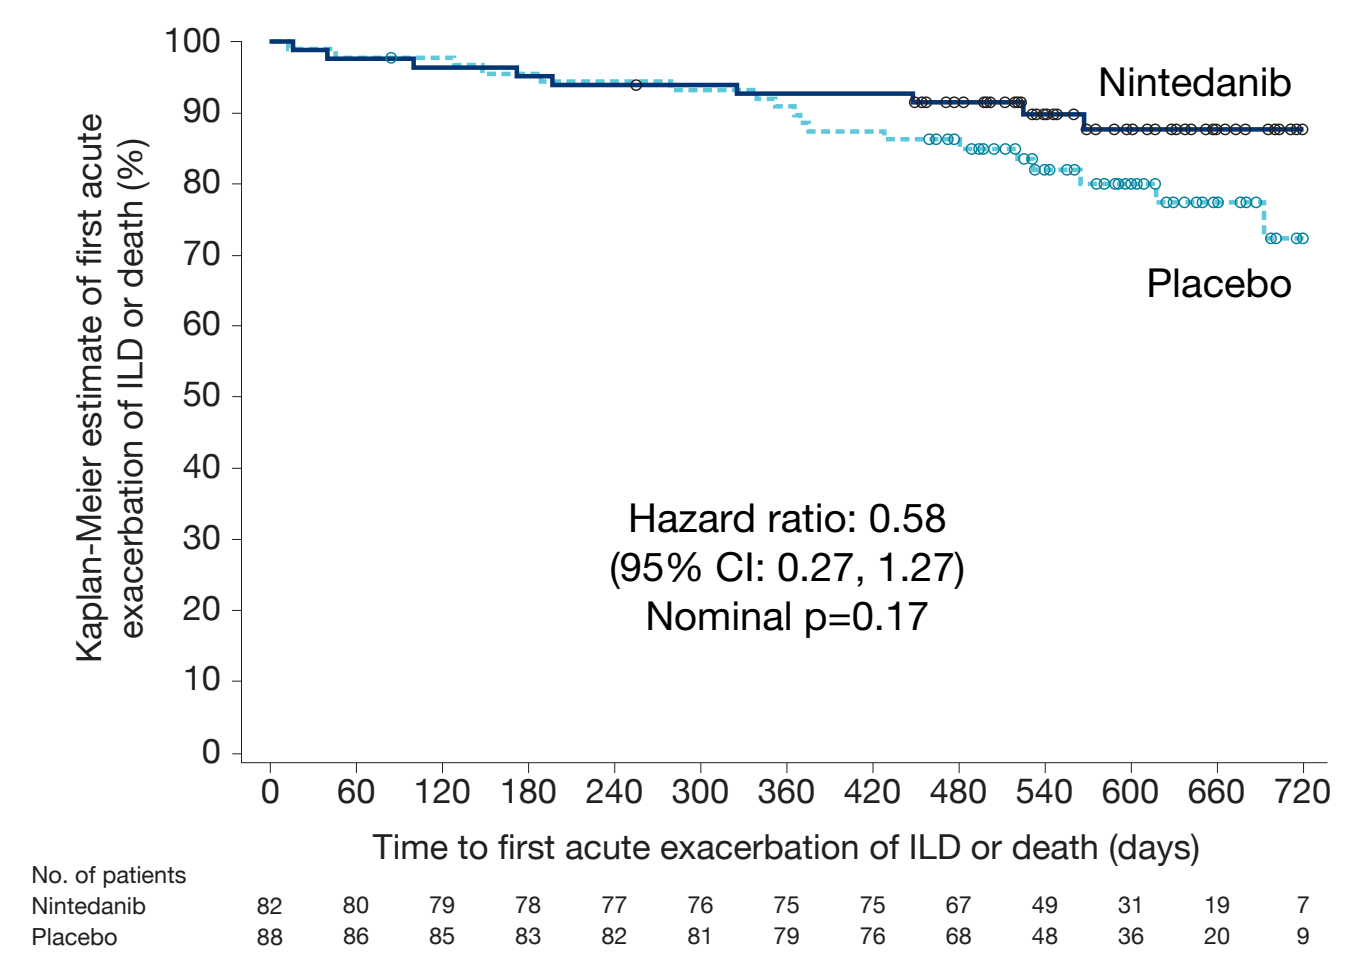
Kaplan-Meier estimate of first acute exacerbation of ILD or death (%)
100 90 80 70 60 50 40 30 20 10 0
Nintedanib
Placebo
Hazard ratio: 0.58
(95% CI: 0.27, 1.27)
Nominal p=0.17
0 60 120 180 240 300 360 420 480 540 600 660 720
Time to first acute exacerbation of ILD or death (days)
No. of patients
Nintedanib 82 80 79 78 77 76 75 75 67 49 31 19 7
Placebo 88 86 85 83 82 81 79 76 68 48 36 20 9

Time to disease progression or death

- 33 patients (40.2%) in the nintedanib group and 47 patients (53.4%) in the placebo group had an absolute decline in FVC ≥10% predicted or died.

Time to disease progression or death

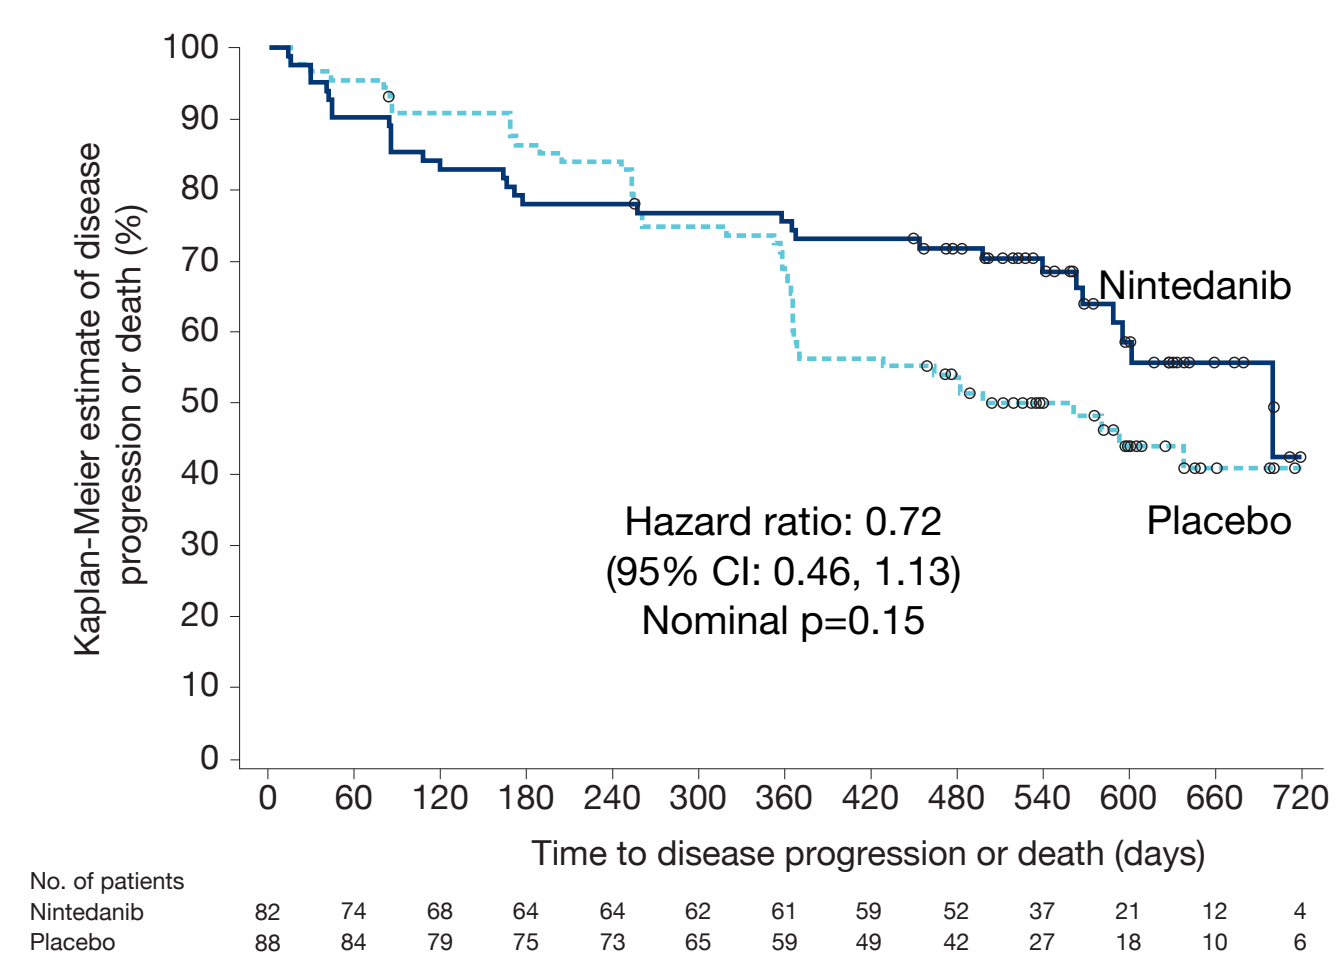
Kaplan-Meier estimate of disease progression or death (%)
100 90 80 70 60 50 40 30 20 10 0
Nintedanib
Placebo
Hazard ratio: 0.72
(95% CI: 0.46, 1.13)
Nominal p=0.15
0 60 120 180 240 300 360 420 480 540 600 660 720
Time to disease progression or death (days)
No. of patients
Nintedanib 82 74 68 64 64 62 61 59 52 37 21 12 4
Placebo 88 84 79 75 73 65 59 49 42 27 18 10 6

Disease progression was defined as an absolute decline in FVC ≥10% predicted.

Adverse events

Most frequent adverse events (reported irrespective of causality)

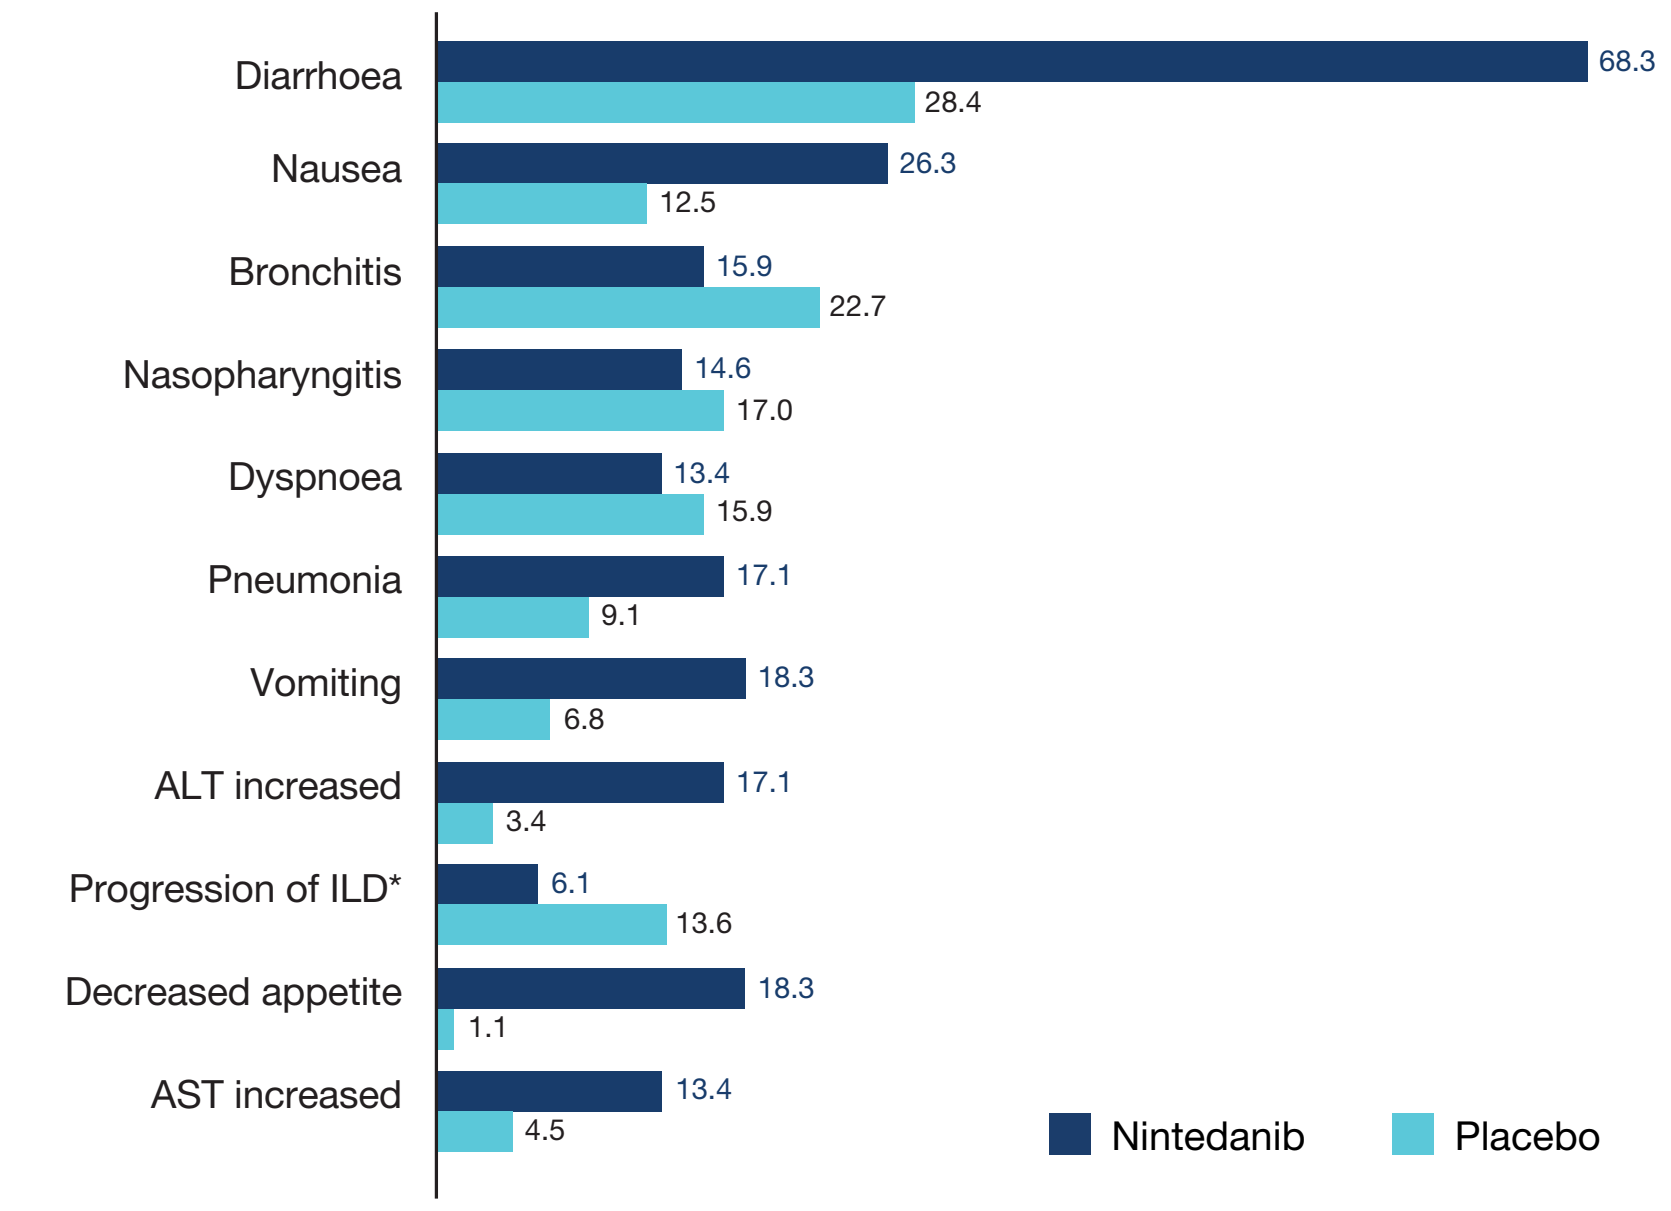
Diarrhoea 68.3 28.4
Nausea 26.3 12.5
Bronchitis 15.9 22.7
Nasopharyngitis 14.6 17.0
Dyspnoea 13.4 15.9
Pneumonia 17.1 9.1
Vomiting 18.3 6.8
ALT increased 17.1 3.4
Progression of ILD* 6.1 13.6
Decreased appetite 18.3 1.1
AST increased 13.4 4.5
Nintedanib
Placebo

Adverse events (reported irrespective of causality) were coded using preferred terms in the Medical Dictionary for Regulatory Activities (MedDRA). Data are % of subjects with ≥1 such adverse event reported between first trial drug intake and 28 days after the last trial drug intake in >10% of patients in either treatment group. *Corresponded to the MedDRA preferred term "interstitial lung disease". ALT, alanine aminotransferase; AST, aspartate aminotransferase.

Adverse events leading to permanent discontinuation of trial drug

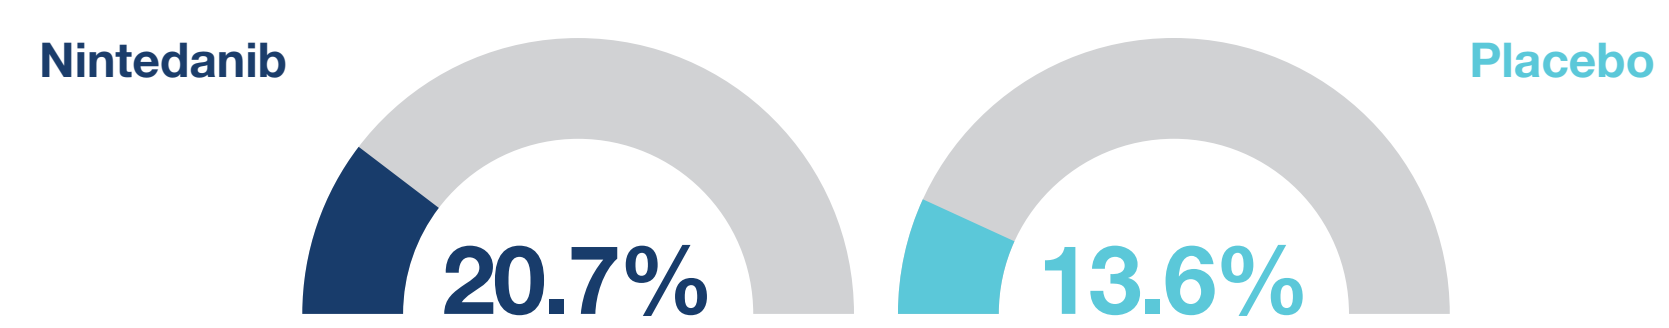
Nintedanib
Placebo
20.7%
13.6%

Data are % of subjects with ≥1 such adverse event.

Serious adverse events

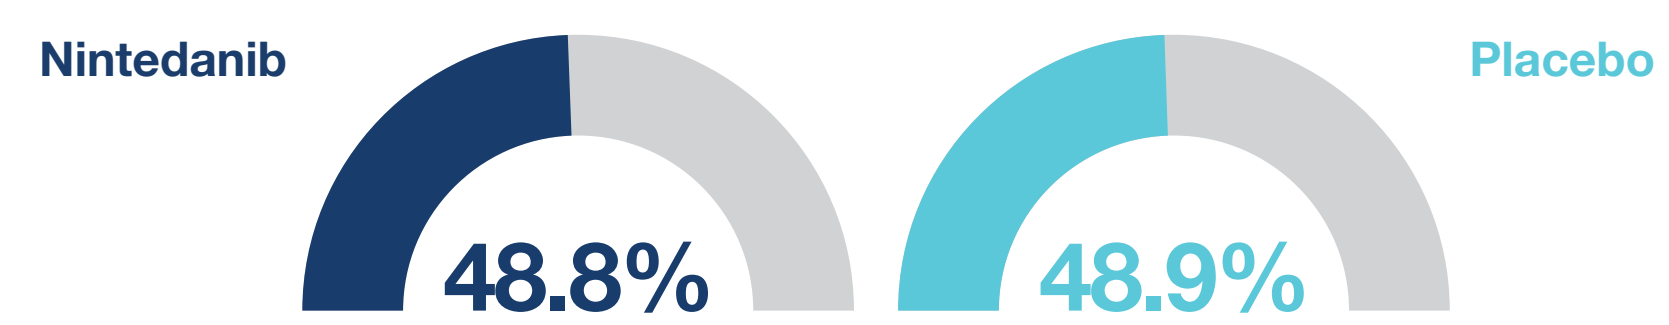
Nintedanib
Placebo
48.8%
48.9%

Data are % of subjects with ≥1 such adverse event reported between first trial drug intake and 28 days after the last trial drug intake. Serious adverse events were defined as events that resulted in death, were life-threatening, resulted in hospitalisation or prolonged hospitalisation, resulted in persistent or clinically significant disability or incapacity, were a congenital anomaly or birth defect, or were deemed serious for any other reason.

Conclusions

- **Data from the INBUILD trial suggest that nintedanib has a clinically meaningful effect on slowing the progression of ILD in patients with chronic fibrosing autoimmune disease-related ILDs with a progressive phenotype, with adverse events that are manageable for most patients.**

References

1. Wollin L, et al. Eur Respir J 2019;54. doi: 10.1183/13993003.00161-2019.
2. Flaherty KR et al. N Engl J Med 2019;381:1718-1727.

Acknowledgements

The INBUILD trial was funded by Boehringer Ingelheim. Editorial and formatting assistance, supported financially by Boehringer Ingelheim, was provided by Elizabeth Ng and Wendy Morris of FleishmanHillard Fishburn, London, UK during preparation of this poster. The authors were fully responsible for all content and editorial decisions, were involved at all stages of poster development and have approved the final version. The authors received no direct compensation related to the development of this poster. Boehringer Ingelheim was given the opportunity to review the poster for medical and scientific accuracy as well as intellectual property considerations.

INTERACTIVE

https://www.usscicomms.com/respiratory/EULAR2020

https://www.usscicomms.com/respiratory/EULAR2020/matteson

Authors' disclosures:

ELM has received a grant from Pfizer and has served as a consultant/speaker for Boehringer Ingelheim (BI), Gilead, TympoBio, Arena Pharmaceuticals and Simply Speaking. CK has served as a consultant/speaker for BI. JHWD reports grants from BI and has served as a consultant/speaker for BI. AMHV reports grants from BI and has served as a consultant/speaker for BI, Actelion, Bayer, GlaxoSmithKline (GSK) and Roche. JRS owns shares in BriaCell, Pacific and has served as a consultant/speaker for Atlantic, Blade, Eicos, Eiger, Indalo, Mitsubishi Tanabe, Xenikos, Bayer, BI, Camurus, Corbus, EMD Serono. SM reports grants from Pfizer and has served as a consultant for Novartis, Abbvie, Pfizer. OD reports grants from Actelion, Bayer, BI, Kymera Therapeutics, Mitsubishi Tanabe and consultancy relationship and/or research funding from Abbvie, Actelion, Acceleron Pharma, Amgen, AnaMar, Baecon Discovery, Blade Therapeutics, Bayer, BI, Catenion, Competitive Corpus, Drug Development International Ltd, CSL Behring, ChemomAb, Ergonex, Galapagos NV, Glenmark Pharmaceuticals, GSK, Horizon (Curzion) Pharmaceuticals, Inventiva, Italfarmaco, iQone, iQvia, Kymera Therapeutics, Lilly, medac, Medscape, Mitsubishi Tanabe Pharma, Merck Sharp & Dohme (MSD), Novartis, Pfizer, Roche, Sanofi, Target BioScience, UCB; he has a patent mir-29 for the treatment of SSc (US8247389, EP2331143). PFD reports grants from Genentech and has served as a consultant for BI. AJ, RSH, SS and MQ are employees of BI. KRF reports grants from BI and has served as a consultant for BI, Veracyte, Genentech, Bellerophon, Blade.

Poster developed for the Annual European Congress of Rheumatology, 2020.