Community advisory board input can make lay summaries of clinical trial results more understandable

Joep Welling,[1] Annelise Roennow,[1,2] Maureen Sauvé,[3] Edith Brown,[4] Ilaria Galetti,[1,5] Alex Gonzalez,[6] Alexandra Paula Portales Guiraud,[7] Ann Tyrrell Kennedy,[1] Catarina Leite,[8] Robert J. Riggs,[9] Alison Zheng,[10] Matea Perkovic Popovic,[11] Annie Gilbert,[12] Lizette Moros,[13] Kamila Sroka-Saidi,[14] Thomas Schindler,[13] Henrik Finnern[13]

[1]Federation of European Scleroderma Associations aisbl. (FESCA), Saint Maur, Belgium; [2]Sklerodermiforeningen, Rødovre, Denmark; [3]Scleroderma Canada, Hamilton, Canada; [4]Scleroderma and Raynaud's UK, London, UK; [5]Gruppo Italiano per la Lotta alla Sclerodermia, Milan, Italy; [6]Scleroderma Research Foundation, San Francisco, USA; [7]Asociación Española de Esclerodermia, Madrid, Spain; [8]Associação Portuguesa de Doentes com Esclerodermia, Matosinhos, Portugal; [9]Scleroderma Foundation, Danvers, MA, USA; [10]Chinese Organization for Scleroderma, China; [11]Croatian Society for Patients with Scleroderma, Zagreb, Croatia; [12]Patient Advocacy Relations Consultant to: [13]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany; [14]Boehringer Ingelheim Pharma GmbH & Co. KG, Biberach, Germany

PARE0009

WHY DO WE NEED LAY SUMMARIES?

A **trial summary for laypersons** must be written for every clinical trial carried out in the EU[1]

The trial sponsor is responsible for writing the lay summary

It must be published within 1 year of study completion

Saved on the EU Clinical Trial Information System, a publicly available database

Lay summaries **share** and **explain** clinical **trial results** in an **easy-to-understand** way for trial participants, patient and caregiver communities, and the general public

Trial sponsors are encouraged to **collaborate** with **patient organisations**, seeking their advice and input to make lay summaries easier to understand and recognising the essential role of patients in clinical research[1–4]

DURING DEVELOPMENT OF THE SENSCIS® TRIAL LAY SUMMARY, ADVICE FROM PATIENT ORGANISATIONS WAS GATHERED THROUGH A COMMUNITY ADVISORY BOARD

The community advisory board was formed based on the EURORDIS charter for collaboration in clinical research,[4] formalised in a co-created memorandum of understanding mutually ratified by patient organisations and the trial sponsor

Over a 7-month period...

...representatives from 11 scleroderma patient organisations covering a range of countries and regions...

...participated in 3 structured meetings to review and advise on lay summary materials being developed

An iterative process was used to gather and collate community advisory board advice to help the trial sponsor prepare effective and meaningful lay summaries that are more understandable and more relevant for patients and the general public

Community advisory board
Provide overall advice on lay summaries

Trial sponsor LSG
Prepare drafts of:
- written lay summary
- lay summary video script

Trial sponsor medical + legal teams
Review and advise

Trial sponsor LSG assess all advice and revise materials

Iterative feedback process conducted over a series of 3 structured meetings

Community advisory board
Review and advise through moderated meetings

Trial sponsor medical, legal and LSG teams
Final approval and sign-off

Upload to trial-sponsored website & publicly accessible trial database

COMMUNITY ADVISORY BOARD ADVICE SUPPORTED DEVELOPMENT OF PATIENT-FOCUSED LAY SUMMARIES

Community advisory board input identified ways to make lay summary materials easier to understand and more relevant

Use neutral colour schemes and icons more representative of scleroderma patients
Changes were made to colour schemes, iconography and layout so lay summaries flowed better, were easier to follow and resonated with audiences

Make lay summaries available in patients' native language(s)
Written and video lay summaries are being translated into all participating country languages

First explain what lay summaries are
A stand-alone video called "What are lay summaries?" was created

Videos are an increasingly important source of information for people with specific health needs
The trial-specific written lay summary was accompanied by a video format lay summary

Some of the language is hard to understand, particularly for people with lower literacy levels
Language was simplified to an appropriate level for a lay audience

Revisions were made to improve clarity of lay summary materials in response to community advisory board advice

Original scene

Include medical images to show what worsening lung fibrosis really looks like

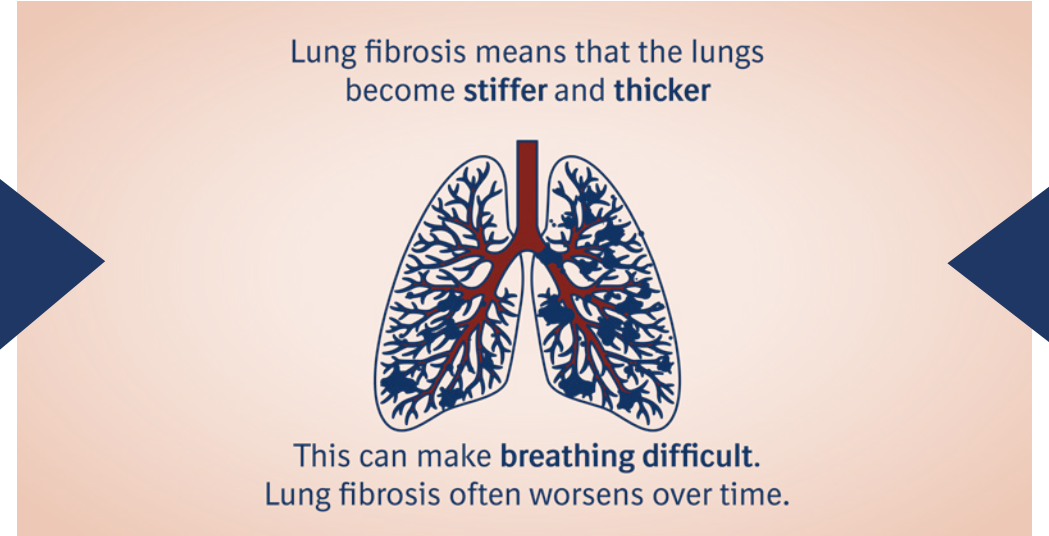
Lung fibrosis means that the lungs become stiffer and thicker
This can make breathing difficult.
Lung fibrosis often worsens over time.

Illustrate what happens to the lungs when fibrosis occurs so it is easier to understand

Revised scene

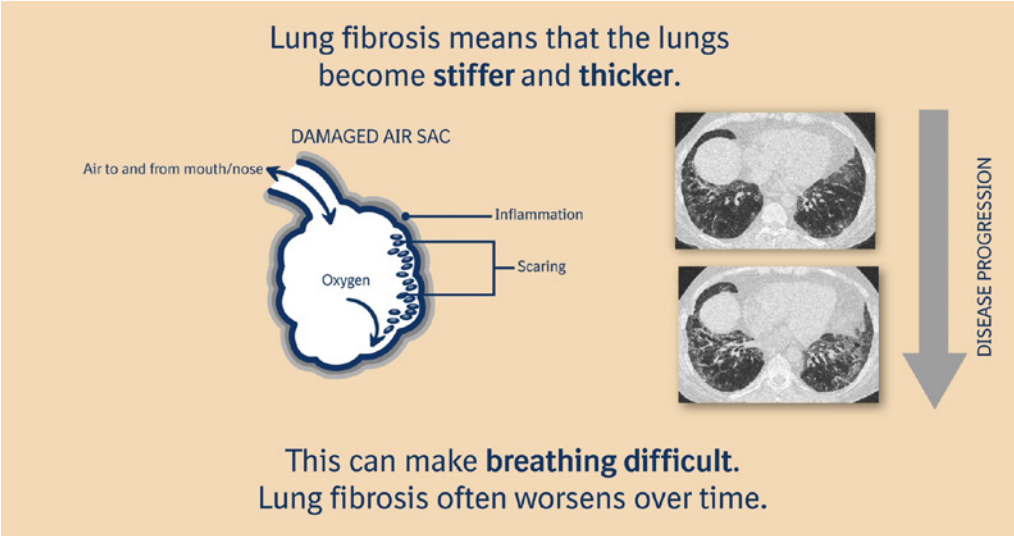
Lung fibrosis means that the lungs become stiffer and thicker.
DAMAGED AIR SAC
Air to and from mouth/nose
Oxygen
Inflammation
Scaring
DISEASE PROGRESSION
This can make breathing difficult.
Lung fibrosis often worsens over time.

The red colour suggests danger and makes you feel anxious; use a neutral colour

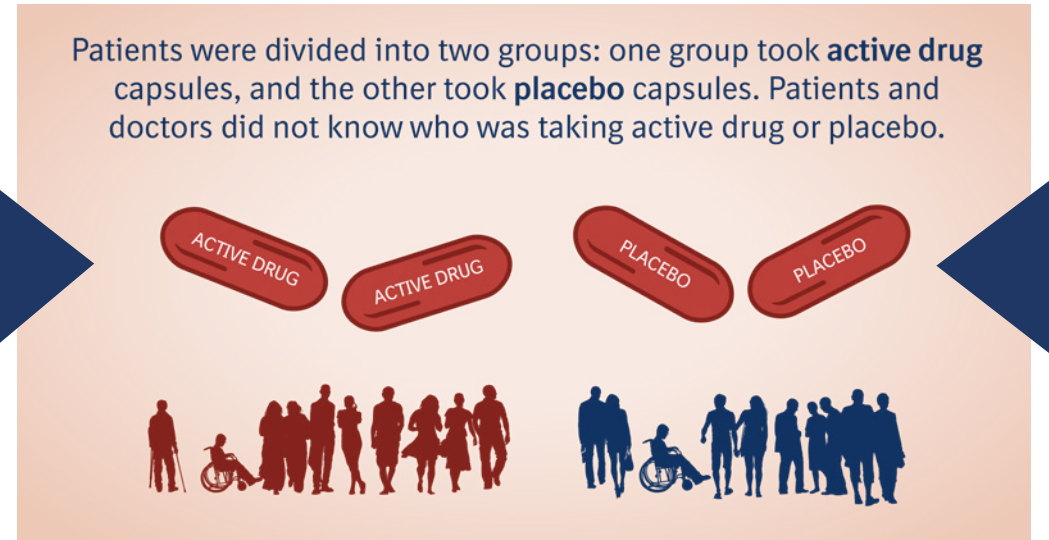
Patients were divided into two groups: one group took active drug capsules, and the other took placebo capsules. Patients and doctors did not know who was taking active drug or placebo.
ACTIVE DRUG
ACTIVE DRUG
PLACEBO
PLACEBO

It looks like the pills are bearing down on the people, like the medicine is more important

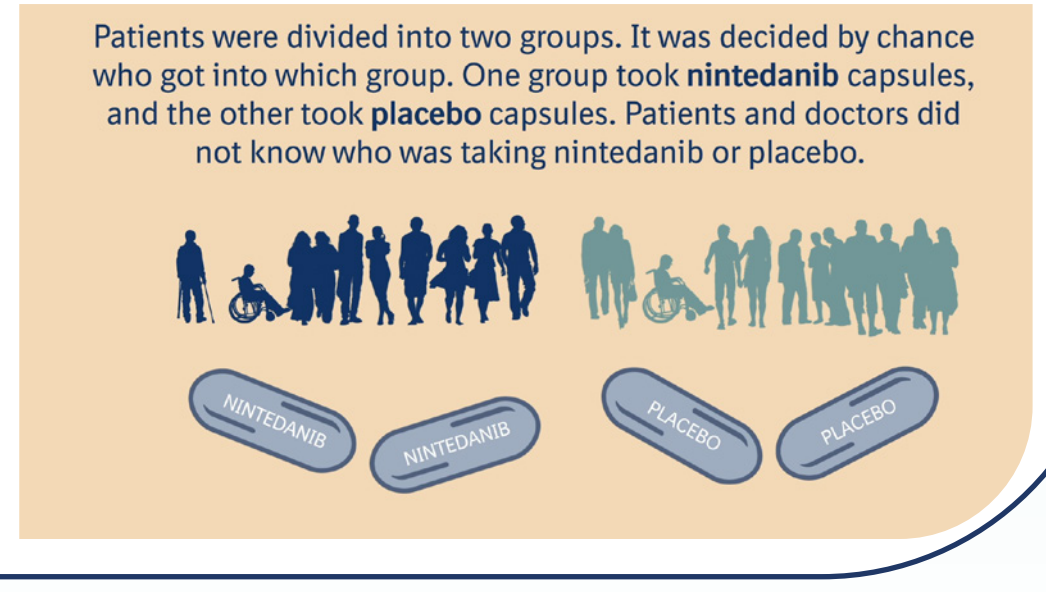
Patients were divided into two groups. It was decided by chance who got into which group. One group took nintedanib capsules, and the other took placebo capsules. Patients and doctors did not know who was taking nintedanib or placebo.
NINTEDANIB
NINTEDANIB
PLACEBO
PLACEBO

CONCLUSION: Structured collection and implementation of community advisory board advice can make lay summary materials more understandable for the patient community, their caregivers and the wider general public

INTERACTIVE

https://www.usscicomms.com/respiratory/EULAR2020

https://www.usscicomms.com/respiratory/EULAR2020/Welling

References

1. EU. Summaries of clinical trial results for laypersons. 2018. Available here: https://ec.europa.eu/health/sites/health/files/files/eudralex/vol10/2017_01_26_summaries_of_ct_results_for_laypersons.pdf;
2. EPF. EPF position: clinical trial results – communication of the lay summary. 2015. Available here: https://www.eu-patient.eu/globalassets/policy/clinicaltrials/epf-lay-summary-position-final_external.pdf;
3. Klingmann I, et al. Front Med 2018; 5:251:e1–11;
4. EURORDIS. Charter for Collaboration in Clinical Research in Rare Diseases. 2009. Available here: https://www.eurordis.org/content/eurordis-charter-clinical-trials-rare-diseases.

Abbreviations

EU, European Union; EURORDIS, Rare Diseases Europe; LSG, lay summary group.

Conflicts of interest

JW Speakers bureau: Four times as a patient advocate for employees of BI Intl and BI MIDI; **MS** Educational grants: BI and Janssen; **AGo** Consultant: Payment made to the patient organisation (Scleroderma Research Foundation) for participation in advisory boards; **AK** Grant/research support: AS FESCA aisbl.; **AZ** Grant/research support: Lorem Vascular, BI China, Jianke Pharmaceutical Co., Ltd., Kangjing Biological Co., Ltd., COFCO Coca-Cola. Consultant: BI. Speakers bureau: BI China; **AGi** Consultant: BI Intl; **LM, TS, HF** Paid employees of BI Intl; **KS-S** Paid employee of BI Pharma; **AR, EB, IG, APPG, CL, RJR** and **MPP:** None declared.

Disclosures

The authors meet criteria for authorship as recommended by the International Committee of Medical Journal Editors (ICMJE). The authors received no direct compensation related to the development of the poster. Medical writing, editorial support and formatting assistance was provided by Caroline Sproat of Nucleus Global, which was contracted and funded by Boehringer Ingelheim International GmbH. Boehringer Ingelheim was given the opportunity to review the poster for medical and scientific accuracy as well as intellectual property considerations.

Poster presented at the European League Against Rheumatism (People with Arthritis and Rheumatism in Europe) E-Congress, 2020.