SAT0329

Is the rate of lung function decline the same in patients with systemic sclerosis-associated ILD (SSc-ILD) who experience weight loss? Data from the SENSCIS® trial

Alain Lescoat,[1] Stéphane Jouneau,[2] Gabriela Riemekasten,[3] Yasuhiro Kondoh,[4] Vanessa Smith,[5] Nina M Patel,[6] John T Huggins,[7] Christian Stock,[8] Martina Gahlemann,[9] Margarida Alves,[10] Christopher P Denton[11] on behalf of the SENSCIS trial investigators

[1]Internal Medicine, CHU South Hospital, Rennes, France; [2]Department of Respiratory Medicine, Competences Centre for Rare Pulmonary Diseases, CHU Rennes, univ Rennes, Rennes, France; [3]University Hospital Charité, Rheumatology and Clinical Immunology, Berlin, and University Hospital Schleswig-Holstein, Rheumatology, Lübeck, Germany; [4]Department of Respiratory Medicine and Allergy, Tosei General Hospital, Japan; [5]Department of Rheumatology, Ghent University Hospital, Ghent, Belgium; Department of Internal Medicine, Ghent University, Ghent, Belgium; [6]Division of Pulmonary, Allergy, and Critical Care Medicine, Columbia University College of Physicians and Surgeons/New York-Presbyterian Hospital, New York, NY, USA; [7]Medical University of South Carolina, Charleston, SC, USA; [8]Boehringer Ingelheim Pharma GmbH & Co. KG, Ingelheim, Germany; [9]Boehringer Ingelheim (Schweiz) GmbH, Basel, Switzerland; [10]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany; [11]University College London Division of Medicine, Centre for Rheumatology and Connective Tissue Diseases, London, UK

Introduction

- Systemic sclerosis (SSc) is commonly associated with gastrointestinal complications, which increase the risk of malabsorption and underweight.[1]
- In the SENSCIS trial in patients with SSc-ILD, nintedanib reduced the rate of decline in forced vital capacity (FVC) (mL/year) over 52 weeks by 44% compared with placebo, with an adverse event profile characterised mainly by gastrointestinal events.[2]

Aim

- To assess the rate of FVC decline and adverse events in subgroups by weight loss ≤5% vs >5% over 52 weeks in the SENSCIS trial.

Methods

Patients

- Eligibility criteria included first non-Raynaud symptom ≤7 years before screening, extent of fibrotic ILD ≥10% on a high-resolution computed tomography (HRCT) scan, FVC ≥40% predicted and diffusion capacity of the lung for carbon monoxide (DLco) 30–89% predicted.
- Patients taking prednisone ≤10 mg/day or equivalent and/or stable therapy with mycophenolate or methotrexate for ≥6 months were allowed to participate.
- Patients were randomised to receive nintedanib or placebo until the last patient had reached week 52 but for ≤100 weeks.

Analyses

- In a non-randomised comparison, we assessed the following in the nintedanib and placebo groups in subgroups by weight loss ≤5% vs >5% over 52 weeks:
 - Rate of decline in FVC (mL/year) over 52 weeks.
 - Proportions of patients who met proposed thresholds for minimal clinically important differences (MCID) for worsened FVC (absolute decrease ≥3.3% predicted) or stable/improved FVC (absolute decrease <3.3% predicted) at week 52, based on estimates derived from Scleroderma Lung Studies I and II, anchored to the health transition question from the Medical Outcomes Short Form-36.[3]
 - Adverse events.

Results

Proportions of patients with weight loss ≤5% and >5% over 52 weeks

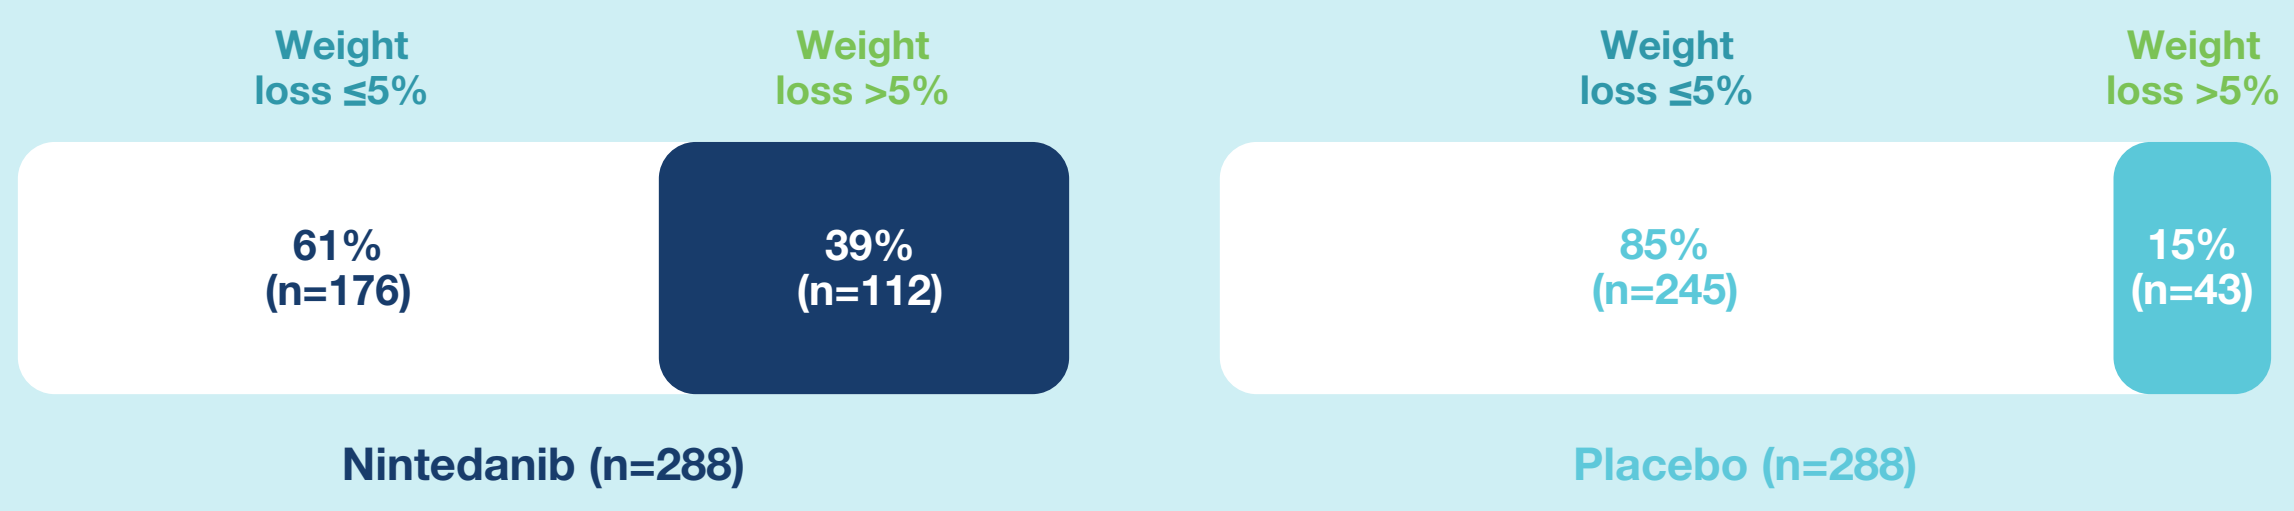

Weight loss ≤5%
Weight loss >5%
61% (n=176)
39% (n=112)
Nintedanib (n=288)
Weight loss ≤5%
Weight loss >5%
85% (n=245)
15% (n=43)
Placebo (n=288)

Baseline characteristics of subgroups by weight loss ≤5% vs >5% over 52 weeks

Weight loss over 52 weeks	≤5%	>5%
Female (%)	72.9	81.3
Age (years)	52.9	57.0
Weight (kg)	69.6	70.1
BMI (kg/m²)	25.7	26.5
Years since onset of non-Raynaud symptom	3.5	3.4
dcSSc (%)	51.3	53.5
mRSS	10.9	11.7
FVC % predicted	73.1	71.0
DLco % predicted	53.3	52.3
Taking mycophenolate (%)	51.8	39.4

Mean or % of patients.
dcSSc, diffuse cutaneous SSc; mRSS, modified Rodnan skin score.

- In both the subgroups by weight loss over 52 weeks, standardised differences between the nintedanib and placebo groups in the baseline values of potential confounders were <0.2 (indicating negligible differences between the nintedanib and placebo groups).

Annual rate of decline in FVC (mL/year)

- In the placebo group, the mean annual rate of decline in FVC was similar between patients who had weight loss ≤5% and >5% over 52 weeks (Figure 1).
- In both the subgroups by weight loss, the estimated annual rate of decline in FVC was lower in patients treated with nintedanib than placebo, with no evidence of heterogeneity between the subgroups (p=0.68 for interaction) (Figure 1).

Figure 1. *Rate of decline in FVC (mL/year) over 52 weeks in subgroups by weight loss over 52 weeks*

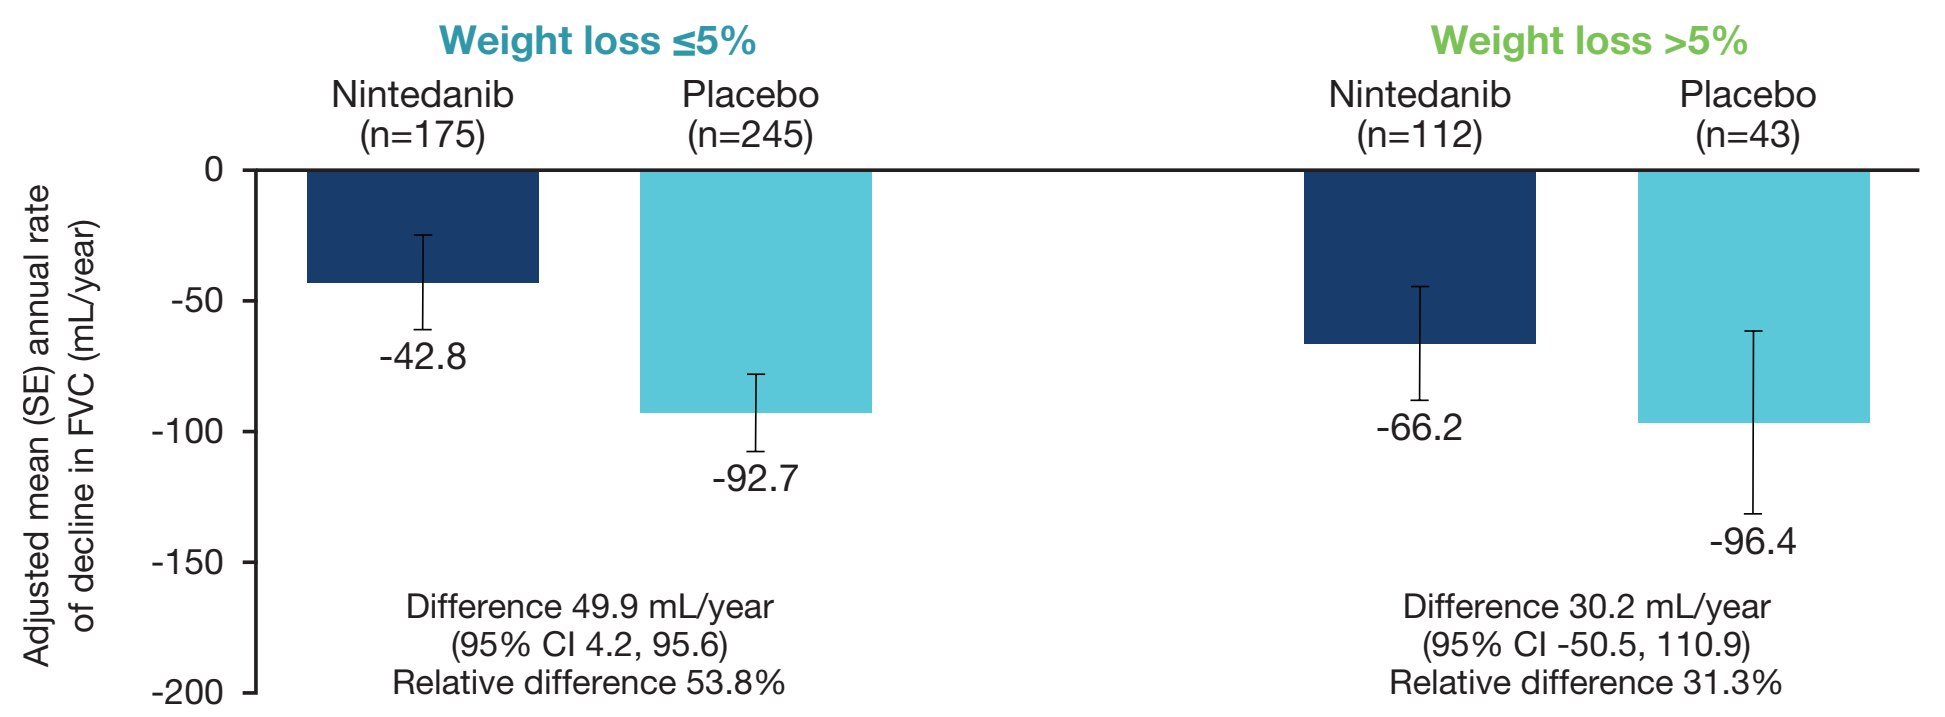

Weight loss ≤5%
Nintedanib (n=175)
Placebo (n=245)
Weight loss >5%
Nintedanib (n=112)
Placebo (n=43)
Adjusted mean (SE) annual rate of decline in FVC (mL/year)
0
-50
-100
-150
-200
-42.8
-92.7
-66.2
-96.4
Difference 49.9 mL/year (95% CI 4.2, 95.6) Relative difference 53.8%
Difference 30.2 mL/year (95% CI -50.5, 110.9) Relative difference 31.3%

Treatment-by-time-by-subgroup interaction p=0.68.

Proportion of patients with stable/improved and worsened FVC

- The proportion of patients with stable/improved FVC was higher, and the proportion with worsened FVC was lower, in patients treated with nintedanib than placebo in both subgroups by weight loss over 52 weeks (Figure 2).

Figure 2. *Proportions of subjects with stable/improved FVC and worsened FVC at week 52 in subgroups by weight loss over 52 weeks*

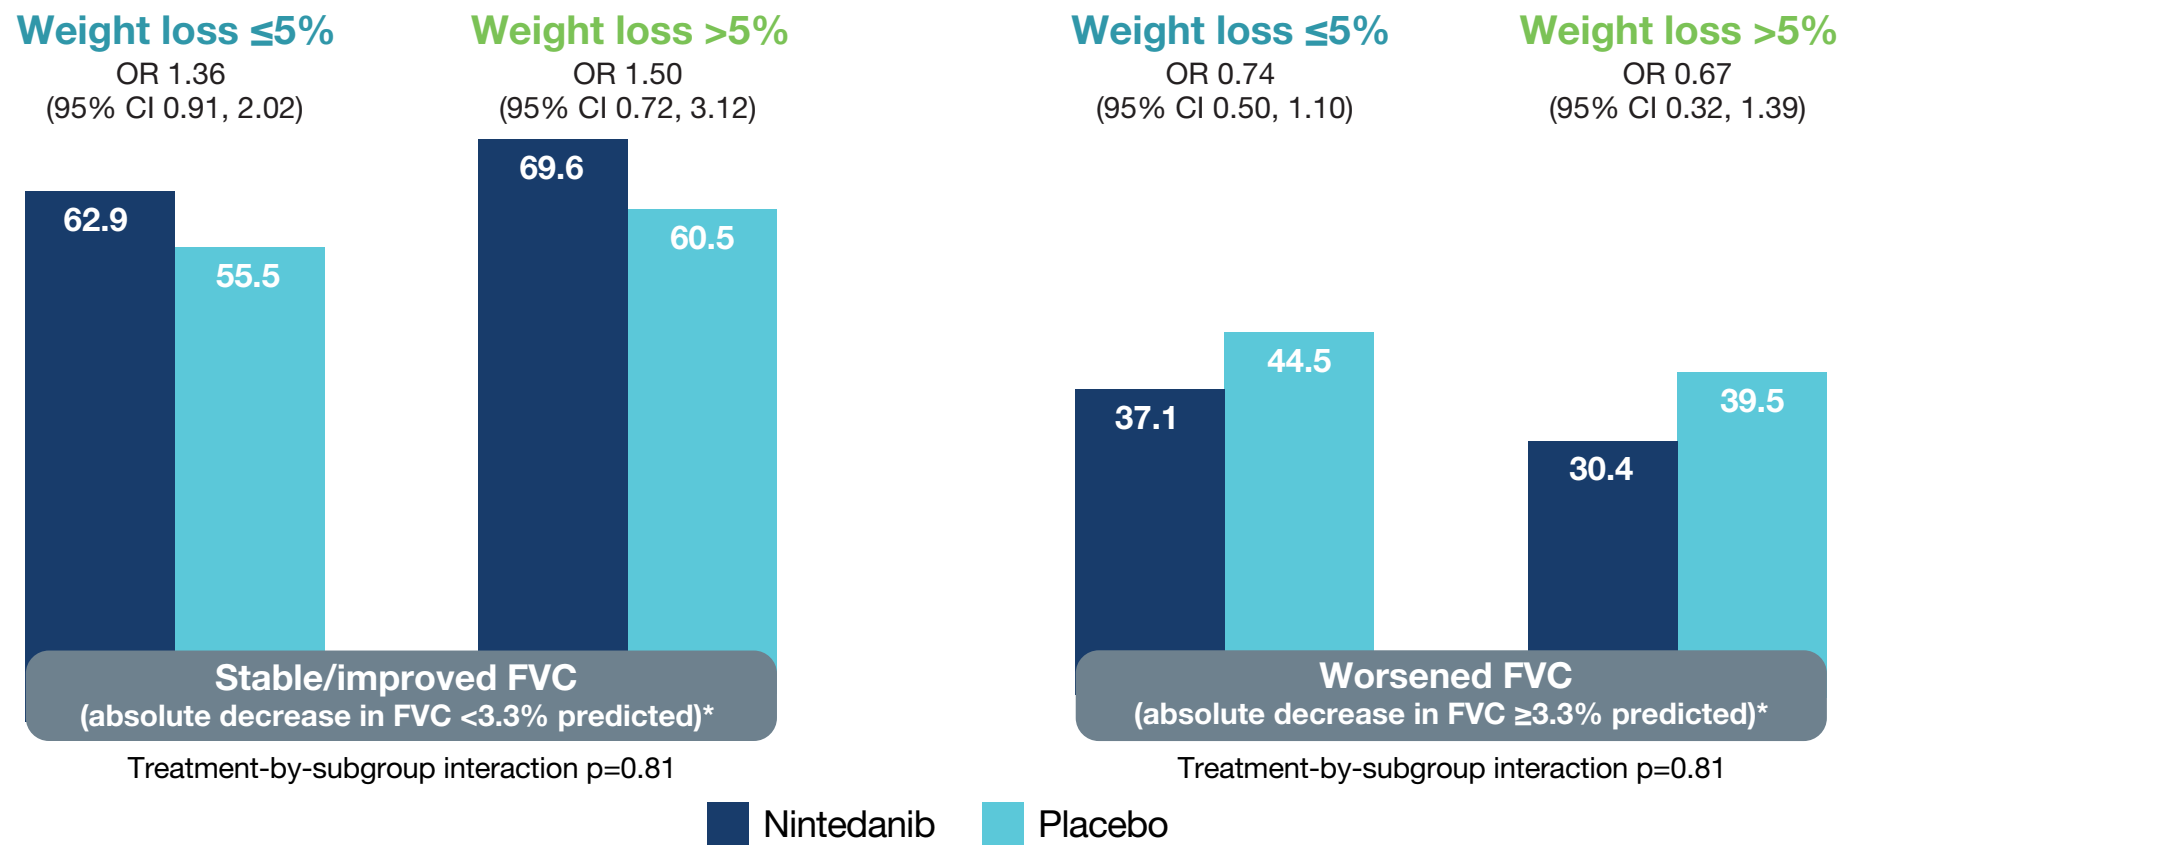

Weight loss ≤5%
OR 1.36 (95% CI 0.91, 2.02)
Weight loss >5%
OR 1.50 (95% CI 0.72, 3.12)
Weight loss ≤5%
OR 0.74 (95% CI 0.50, 1.10)
Weight loss >5%
OR 0.67 (95% CI 0.32, 1.39)
62.9
55.5
69.6
60.5
37.1
44.5
30.4
39.5
Stable/improved FVC (absolute decrease in FVC <3.3% predicted)*
Worsened FVC (absolute decrease in FVC ≥3.3% predicted)*
Treatment-by-subgroup interaction p=0.81
Treatment-by-subgroup interaction p=0.81
Nintedanib
Placebo

*Based on estimates derived from Scleroderma Lung Studies I and II, anchored to the health transition question from the Medical Outcomes Short Form-36.[3]
Missing values were imputed using a worst value carried forward approach.

Adverse events

- Nausea and vomiting (but not diarrhoea) were reported more frequently in patients with weight loss >5% than ≤5% over 52 weeks (Figure 3).

Figure 3. *Most frequent adverse events in subgroups by weight loss over 52 weeks*

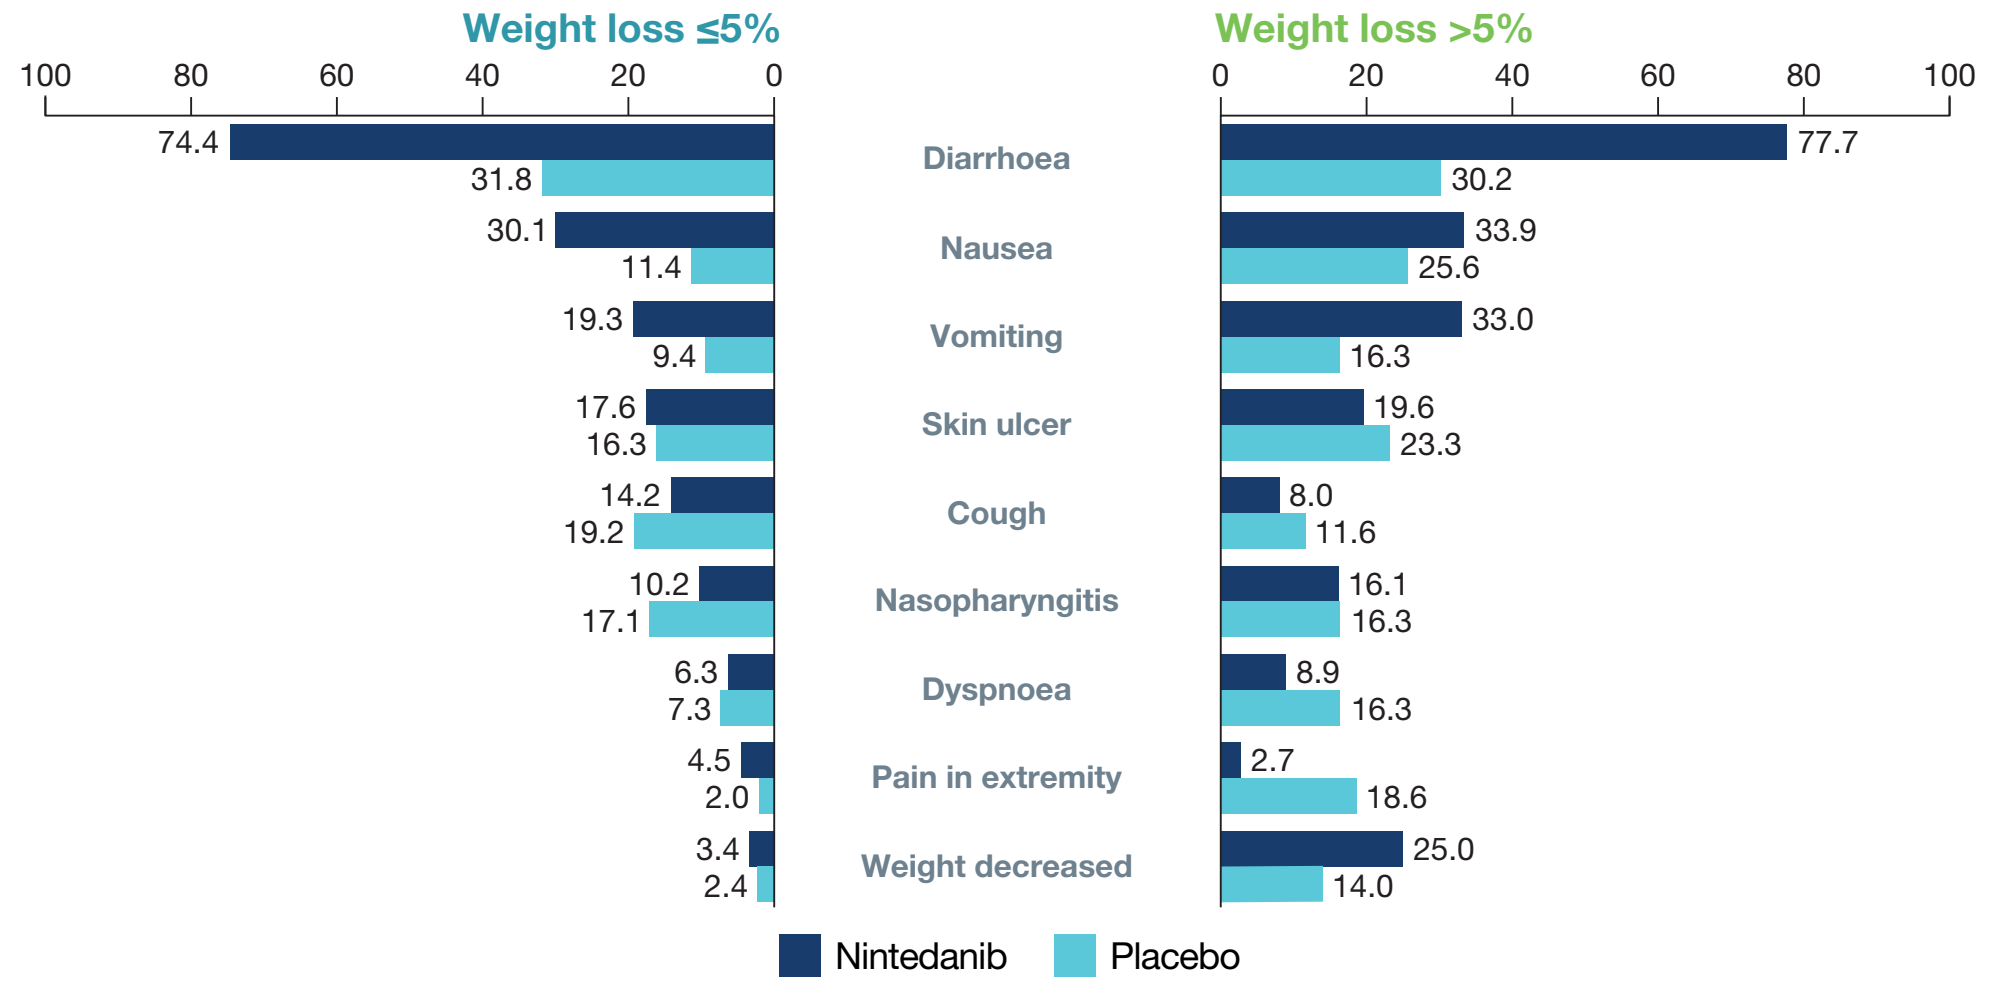

Weight loss ≤5%
Weight loss >5%
100 80 60 40 20 0
0 20 40 60 80 100
Diarrhoea 74.4 31.8 | 77.7 30.2
Nausea 30.1 11.4 | 33.9 25.6
Vomiting 19.3 9.4 | 33.0 16.3
Skin ulcer 17.6 16.3 | 19.6 23.3
Cough 14.2 19.2 | 8.0 11.6
Nasopharyngitis 10.2 17.1 | 16.1 16.3
Dyspnoea 6.3 7.3 | 8.9 16.3
Pain in extremity 4.5 2.0 | 2.7 18.6
Weight decreased 3.4 2.4 | 25.0 14.0
Nintedanib
Placebo

Adverse events reported (irrespective of causality) were coded using preferred terms in the Medical Dictionary for Regulatory Activities (MedDRA). Data shown are % of patients with ≥1 such adverse event, reported over 52 weeks (or until 28 days after last trial drug intake in patients who discontinued trial drug before week 52). Adverse events reported by >15% of patients in any subgroup are shown.

- Adverse events leading to treatment discontinuation occurred at a similar frequency in the subgroups with weight loss ≤5% and >5% over 52 weeks (Figure 4).

Figure 4. *Proportions of patients with adverse events leading to treatment discontinuation in subgroups by weight loss over 52 weeks*

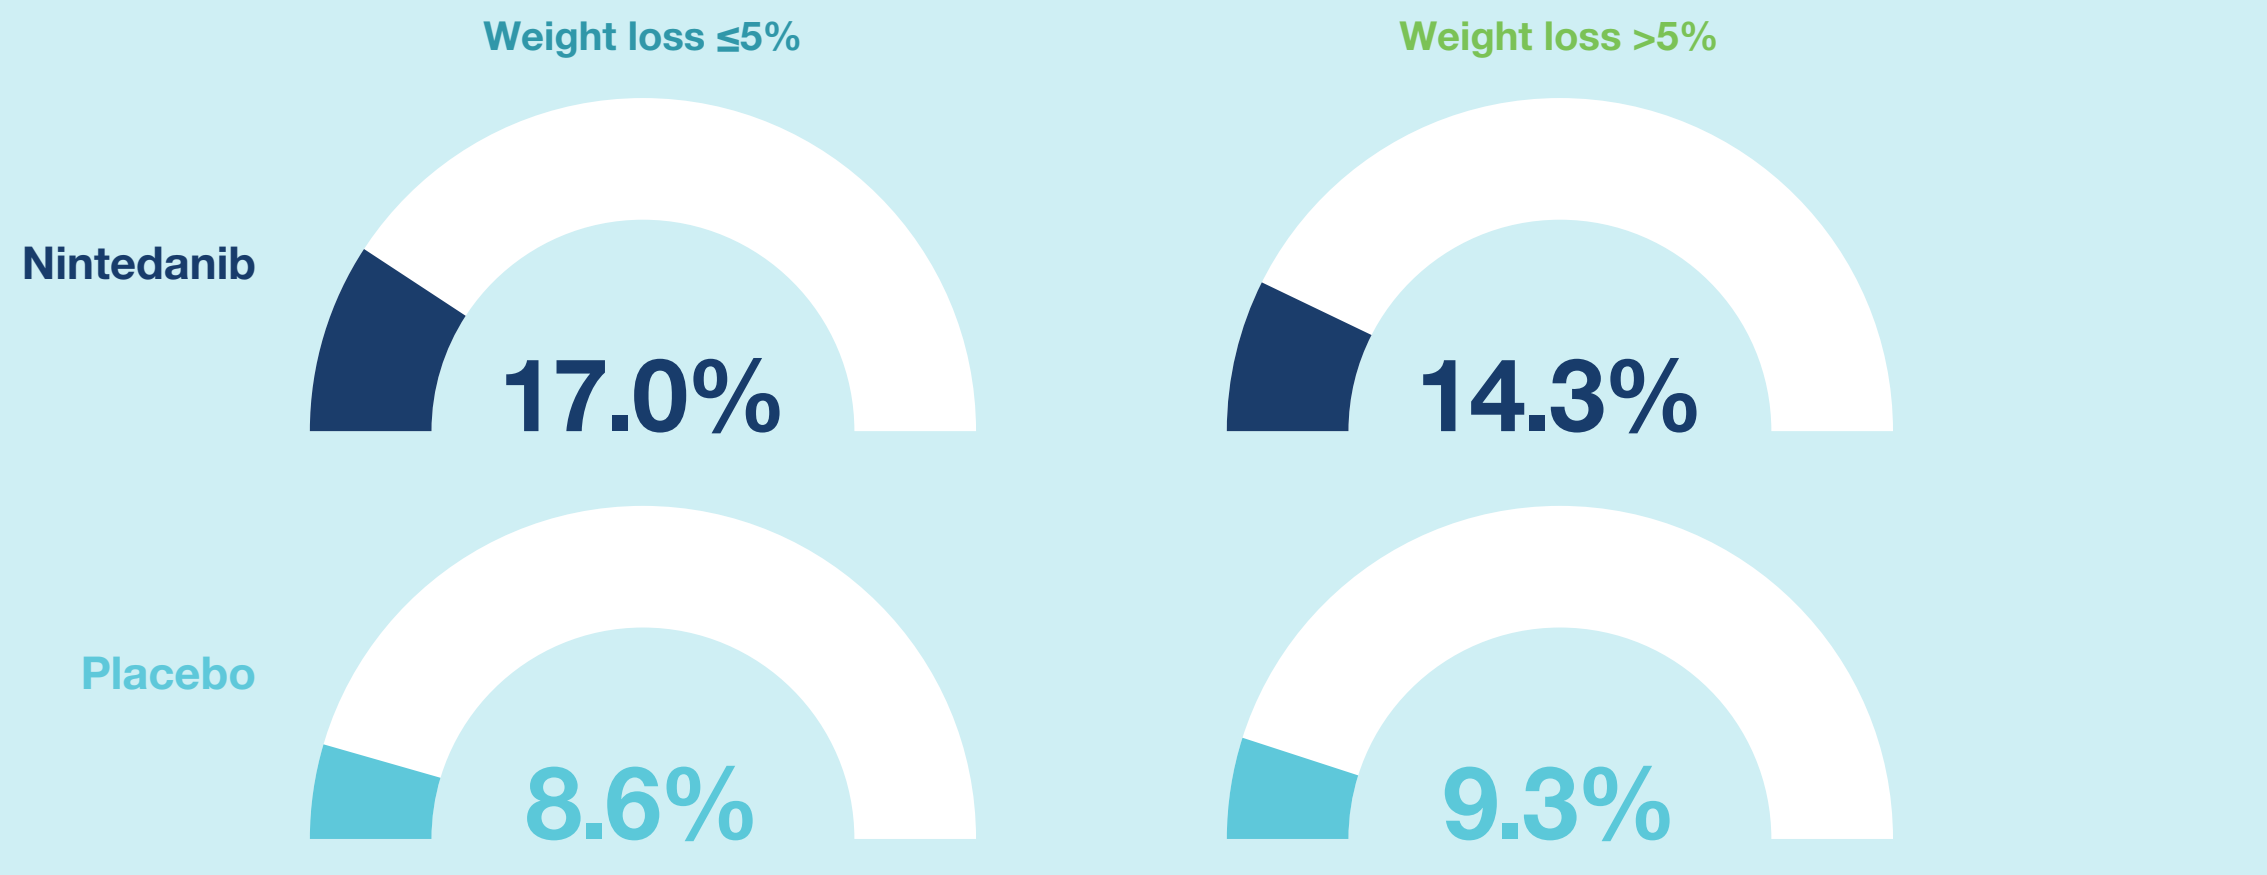

Weight loss ≤5%
Weight loss >5%
Nintedanib
17.0%
14.3%
Placebo
8.6%
9.3%

Data are % of subjects with ≥1 such adverse event, reported over 52 weeks (or until 28 days after last trial drug intake in patients who discontinued trial drug before week 52).

- In both subgroups, serious adverse events were more common in patients with weight loss >5% than ≤5% over 52 weeks (Figure 5).

Figure 5. *Proportions of patients with serious adverse events in subgroups by weight loss over 52 weeks*

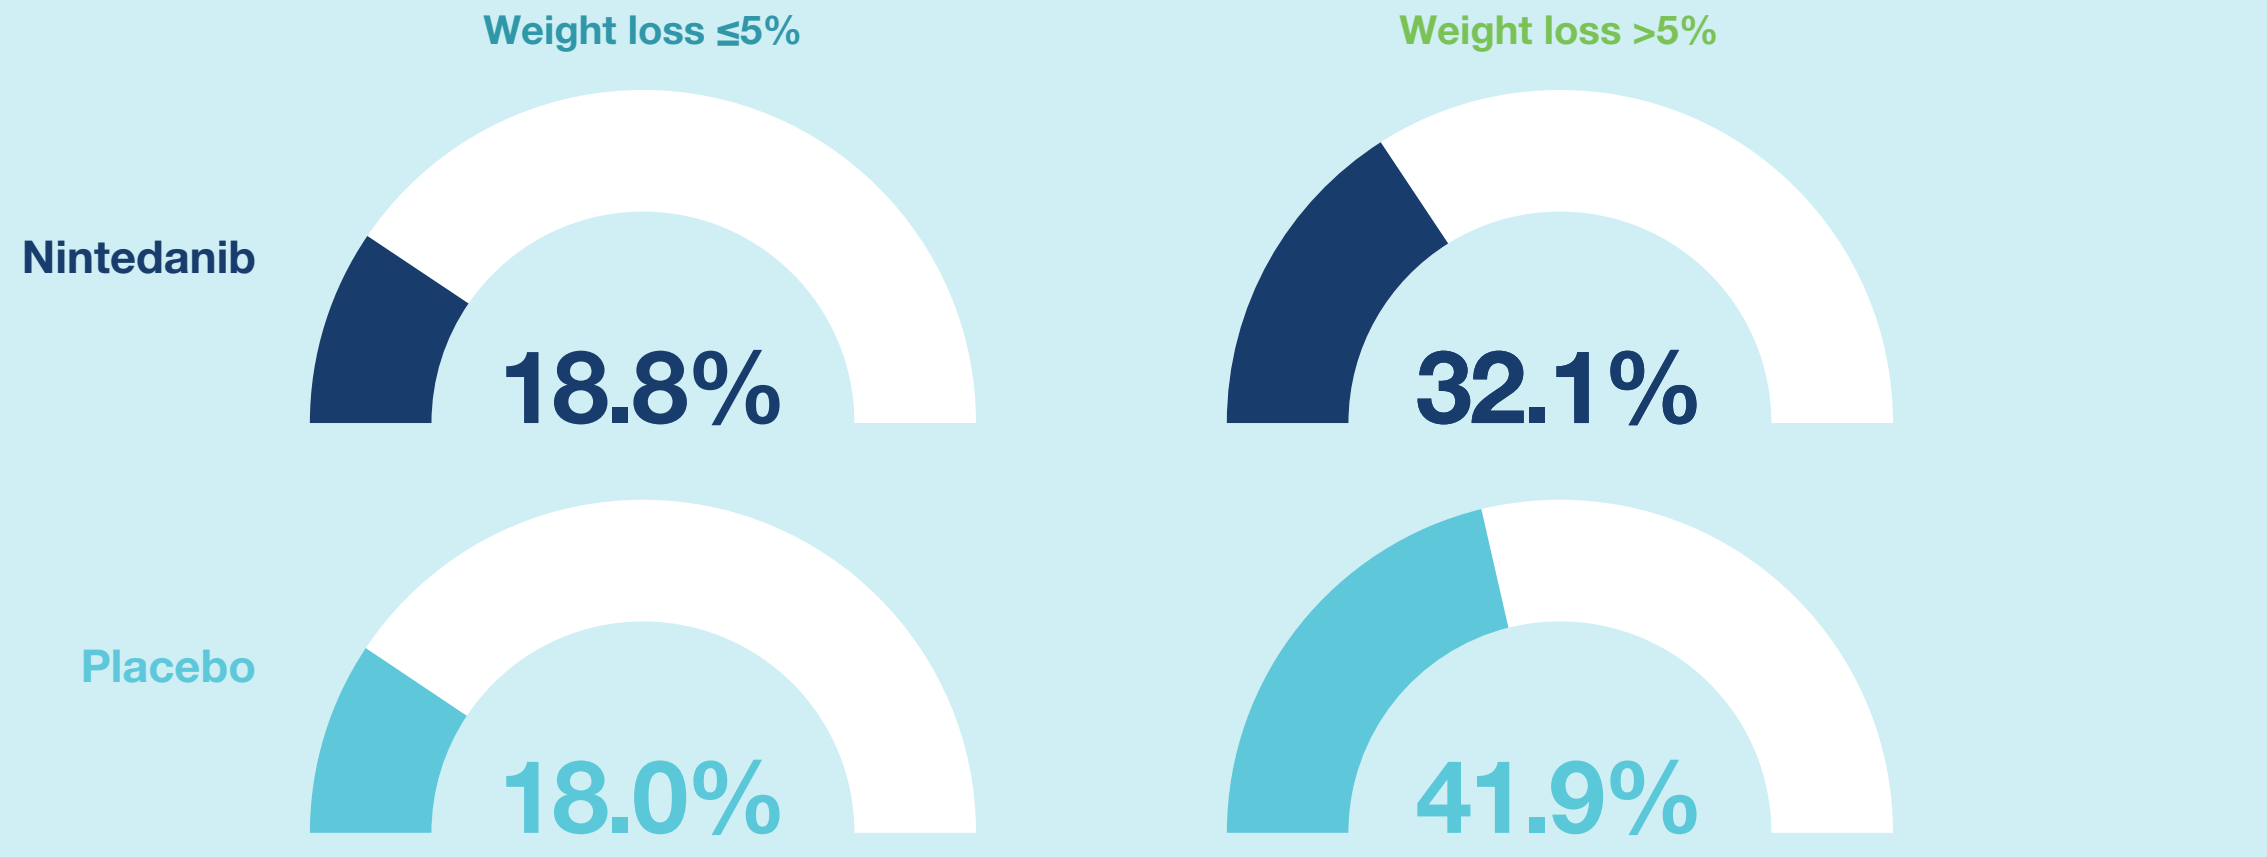

Weight loss ≤5%
Weight loss >5%
Nintedanib
18.8%
32.1%
Placebo
18.0%
41.9%

A serious adverse event was defined as one that resulted in death, was life-threatening, resulted in hospitalisation or prolonged hospitalisation, resulted in persistent or clinically significant disability or incapacity, was a congenital anomaly or birth defect, or was deemed serious for any other reason. Data are % of subjects with ≥1 such adverse event, reported over 52 weeks (or until 28 days after last trial drug intake in patients who discontinued trial drug before week 52).

Conclusions

- **In the SENSCIS trial in patients with SSc-ILD, a greater proportion of patients treated with nintedanib than placebo had weight loss >5% over 52 weeks.**
- **Nintedanib had a consistent effect in reducing the rate of decline in FVC in patients with weight loss ≤5% and >5% over 52 weeks.**
- **Adverse events leading to discontinuation of nintedanib did not occur more frequently in patients with weight loss >5% vs ≤5% over 52 weeks.**

References

1. Forbes A and Marie I. Rheumatology (Oxford) 2009;48:iii36–9.
2. Distler O et al. N Engl J Med 2019;380:2518–28.
3. Kafaja S et al. Am J Respir Crit Care Med 2018;197:644–52.

Acknowledgements

The SENSCIS trial was funded by Boehringer Ingelheim. Editorial and formatting assistance, supported financially by Boehringer Ingelheim, was provided by Elizabeth Ng and Wendy Morris of FleishmanHillard Fishburn, London, UK during preparation of this poster. The authors were fully responsible for all content and editorial decisions, were involved at all stages of poster development and have approved the final version. The authors received no direct compensation related to development of this poster. Boehringer Ingelheim was given the opportunity to review the poster for medical and scientific accuracy as well as intellectual property considerations.

INTERACTIVE

https://www.usscicomms.com/respiratory/EULAR2020

https://www.usscicomms.com/respiratory/EULAR2020/lescoat

Authors' disclosures:
AL has nothing to disclose. SJ reports grants/research support from AIRB, Bellerophon Therapeutics, Biogen, Boehringer Ingelheim (BI), FibroGen, Galecto, Gilead, Pharm-Olam, Pliant, Savara, LVL, Novartis, Roche and has served as a consultant for BI, Novartis, Roche, Actelion, AIRB, AstraZeneca (AZ), Bristol-Myers Squibb (BMS), Chiesi, Genzyme, GlaxoSmithKline (GSK), LVL, Mundipharma, Novartis, Pfizer, Roche, Sanofi. GR has served as a consultant/speaker for CellTrend, Janssen, Actelion, BI, Novartis, Roche, GSK, Pfizer. YK has served as a consultant/speaker for BI, Asahi Kasei, Janssen, Shionogi, Eisai, KYORIN, Mitsubishi Tanabe, Novartis. VS reports grants from BI and Janssen and has served as a consultant/speaker for BI, Actelion, UCB. NMP reports grants from BI and has served as a consultant/speaker for BI and Genentech. JTH has served as a consultant and site investigator for BI. CS, MG and MA are employees of BI. CPD reports grants from GSK, CSL Behring, Inventiva and has served as a consultant for Medscape, Genentech, Actelion, GSK, Sanofi, Inventiva, CSL Behring, BI, Corbus, Acceleron, Curzion, Bayer.

Boehringer Ingelheim

Poster developed for the Annual European Congress of Rheumatology, 2020.