Estimating long-term survival in progressive fibrosing interstitial lung disease (PF-ILD) other than IPF using matched IPF data

Toby M. Maher,[1] Bryony Langford,[2] Alex Diamantopoulos,[2] Klaus B. Rohr,[3] Michael Baldwin,[3] Yoshikazu Inoue[4]

[1]Keck Medicine of University of Southern California, Los Angeles, CA, USA; [2]Symmetron Limited, London, UK; [3]Boehringer Ingelheim International GmbH, Ingelheim am Rhein, Germany; [4]Clinical Research Center, National Hospital Organization Kinki-Chuo Chest Medical Center, Osaka, Japan

BACKGROUND

- There is a lack of survival data from clinical trials and real-world evidence in ILD with a progressive fibrosing phenotype other than IPF (non-IPF PF-ILD).
- The Phase III INBUILD trial showed that nintedanib slows disease progression in non-IPF PF-ILD. [1]
- Since IPF has a similar disease course to PF-ILD, data from long-term IPF studies may be suitable for informing estimates of PF-ILD survival.[2,3]

AIM

- To develop models of long-term survival in patients with PF-ILD in the INBUILD trial using propensity score-matched long-term IPF data.

METHODS

- Propensity score matching was used to weight and match patients with IPF from four nintedanib trials (TOMORROW[4], INPULSIS 1[5] and 2[5], and the long-term extension INPULSIS-ON[6] trial) to patients with PF-ILD in the INBUILD trial.
- Seven traditional survival models were fitted to the matched, weighted IPF data and the three best-fitting models, according to standard statistical tests, were used to generate informative priors for the shape parameter of the Bayesian model.
- These three distributions were then fitted to the PF-ILD data and extrapolated over the long-term.

Methods: preparation, matching and survival analysis

1. Preparation
Database cleaning and study linking

2. Matching
Patients with PF-ILD propensity score-matched (by age, gender, race, FVC, DLco and smoking status) to patients with IPF

3. IPF survival analysis
Traditional survival analysis used to fit models to matched IPF data; three best-fit models selected and used to generate Bayesian priors

4. Bayesian survival analysis
Priors informed by IPF analysis used to model PF-ILD data in Bayesian framework

Patients with IPF
TOMORROW[4], INPULSIS-1[5], INPULSIS-2[5], INPULSIS-ON[6]

Combined before matching → Combined after matching → Dataset used to inform prior in Bayesian survival analysis

Patients with PF-ILD
INBUILD[1]

Dataset before matching → Dataset after matching → Dataset used as evidence in Bayesian survival analysis

Bayesian analyses informed by IPF trial data can be used to extrapolate survival estimates in patients with non-IPF PF-ILD

CONCLUSION

- Using Bayesian analyses informed by IPF trial data provides robust 5-year survival estimates for patients with non-IPF PF-ILD treated with nintedanib or placebo.
- Our results are consistent with clinical experience and real-world evidence from patients with IPF.[7–9]
- These estimates can be used to inform future decision-making in the absence of long-term PF-ILD data.

RESULTS

Gamma, log-logistic and Weibull survival models were the best fit for the matched IPF data

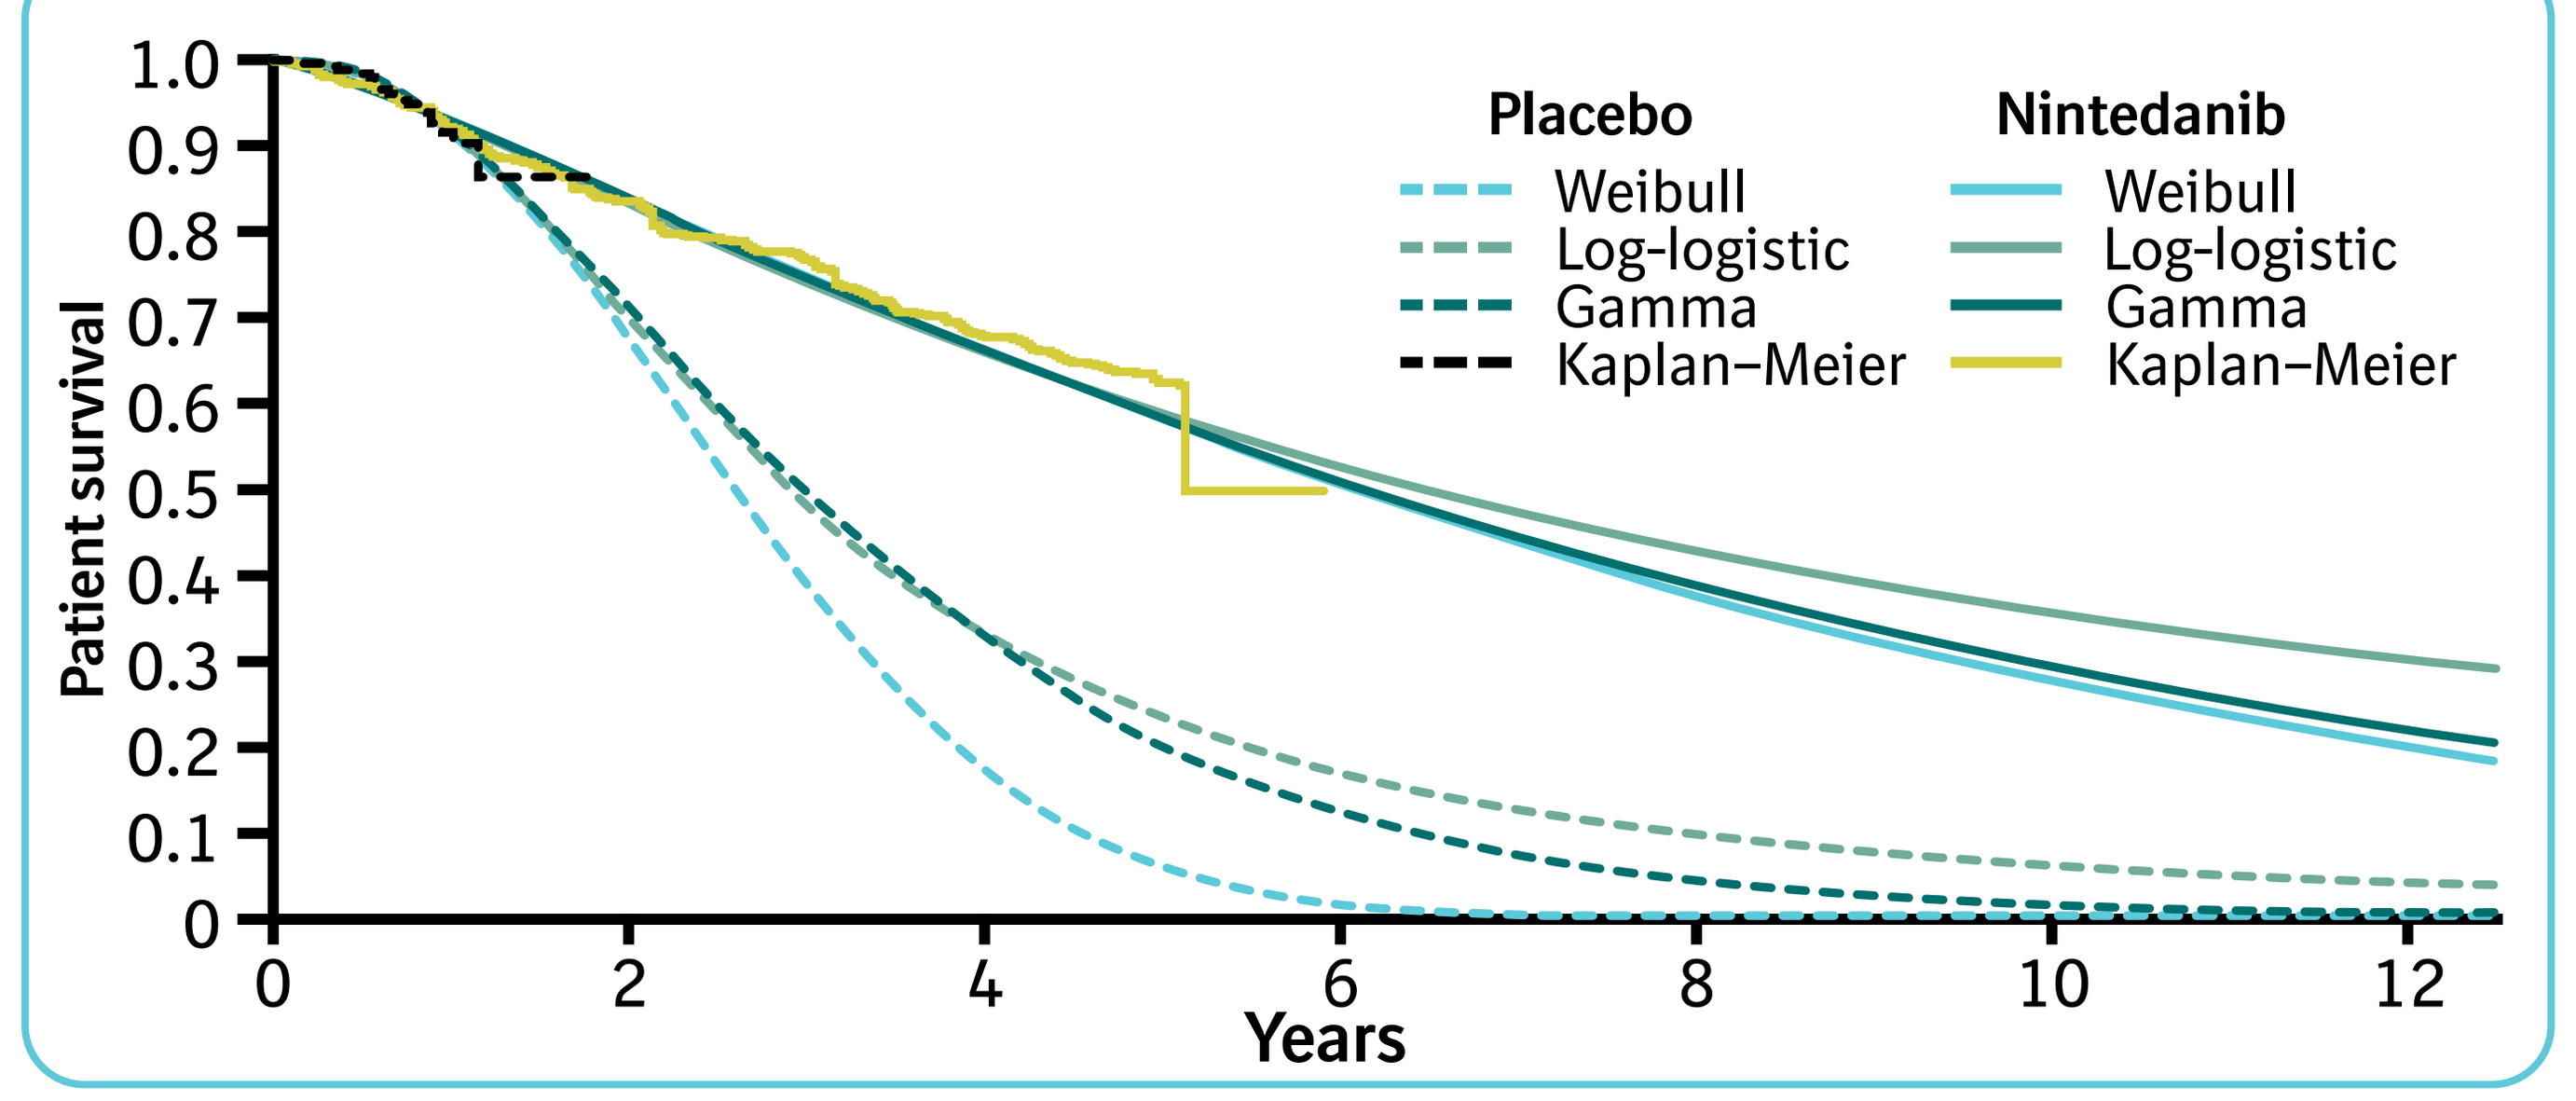
Placebo
Weibull
Log-logistic
Gamma
Kaplan–Meier
Nintedanib
Weibull
Log-logistic
Gamma
Kaplan–Meier
Patient survival
Years

Survival estimates for matched IPF data

	Median OS (years)	
	Nintedanib	Placebo
Gamma	6.13	2.93
Log-logistic	6.48	3.00
Weibull	6.06	2.60

OS estimates for PF-ILD from the Bayesian survival models

Distribution	Median OS (years)		5-year survival (%)	
	Nintedanib	Placebo	Nintedanib	Placebo
Gamma	6.50	3.76	60	32
Log-logistic	6.34	3.73	59	34
Weibull	6.45	3.42	60	21

Nintedanib
Median OS and 5-year survival estimates were consistent across the three models

Placebo
The Weibull model gave a lower estimate of 5-year survival than the other models

Traditional survival analysis results (frequentist approach using INBUILD data alone)

Median OS (years)					
Distribution	Nintedanib	Placebo	**Distribution**	Nintedanib	Placebo
Exponential	11.91	8.46	**Log-logistic**	8.46	3.70
GenGamma	5.75	3.12	**Log-normal**	16.67	5.17
Gompertz	3.94	2.63	**Weibull**	6.90	3.37

- Compared with survival estimates based on INBUILD data only, the Bayesian estimates of median OS in PF-ILD provided less variation and uncertainty.

Bayesian survival analysis using:

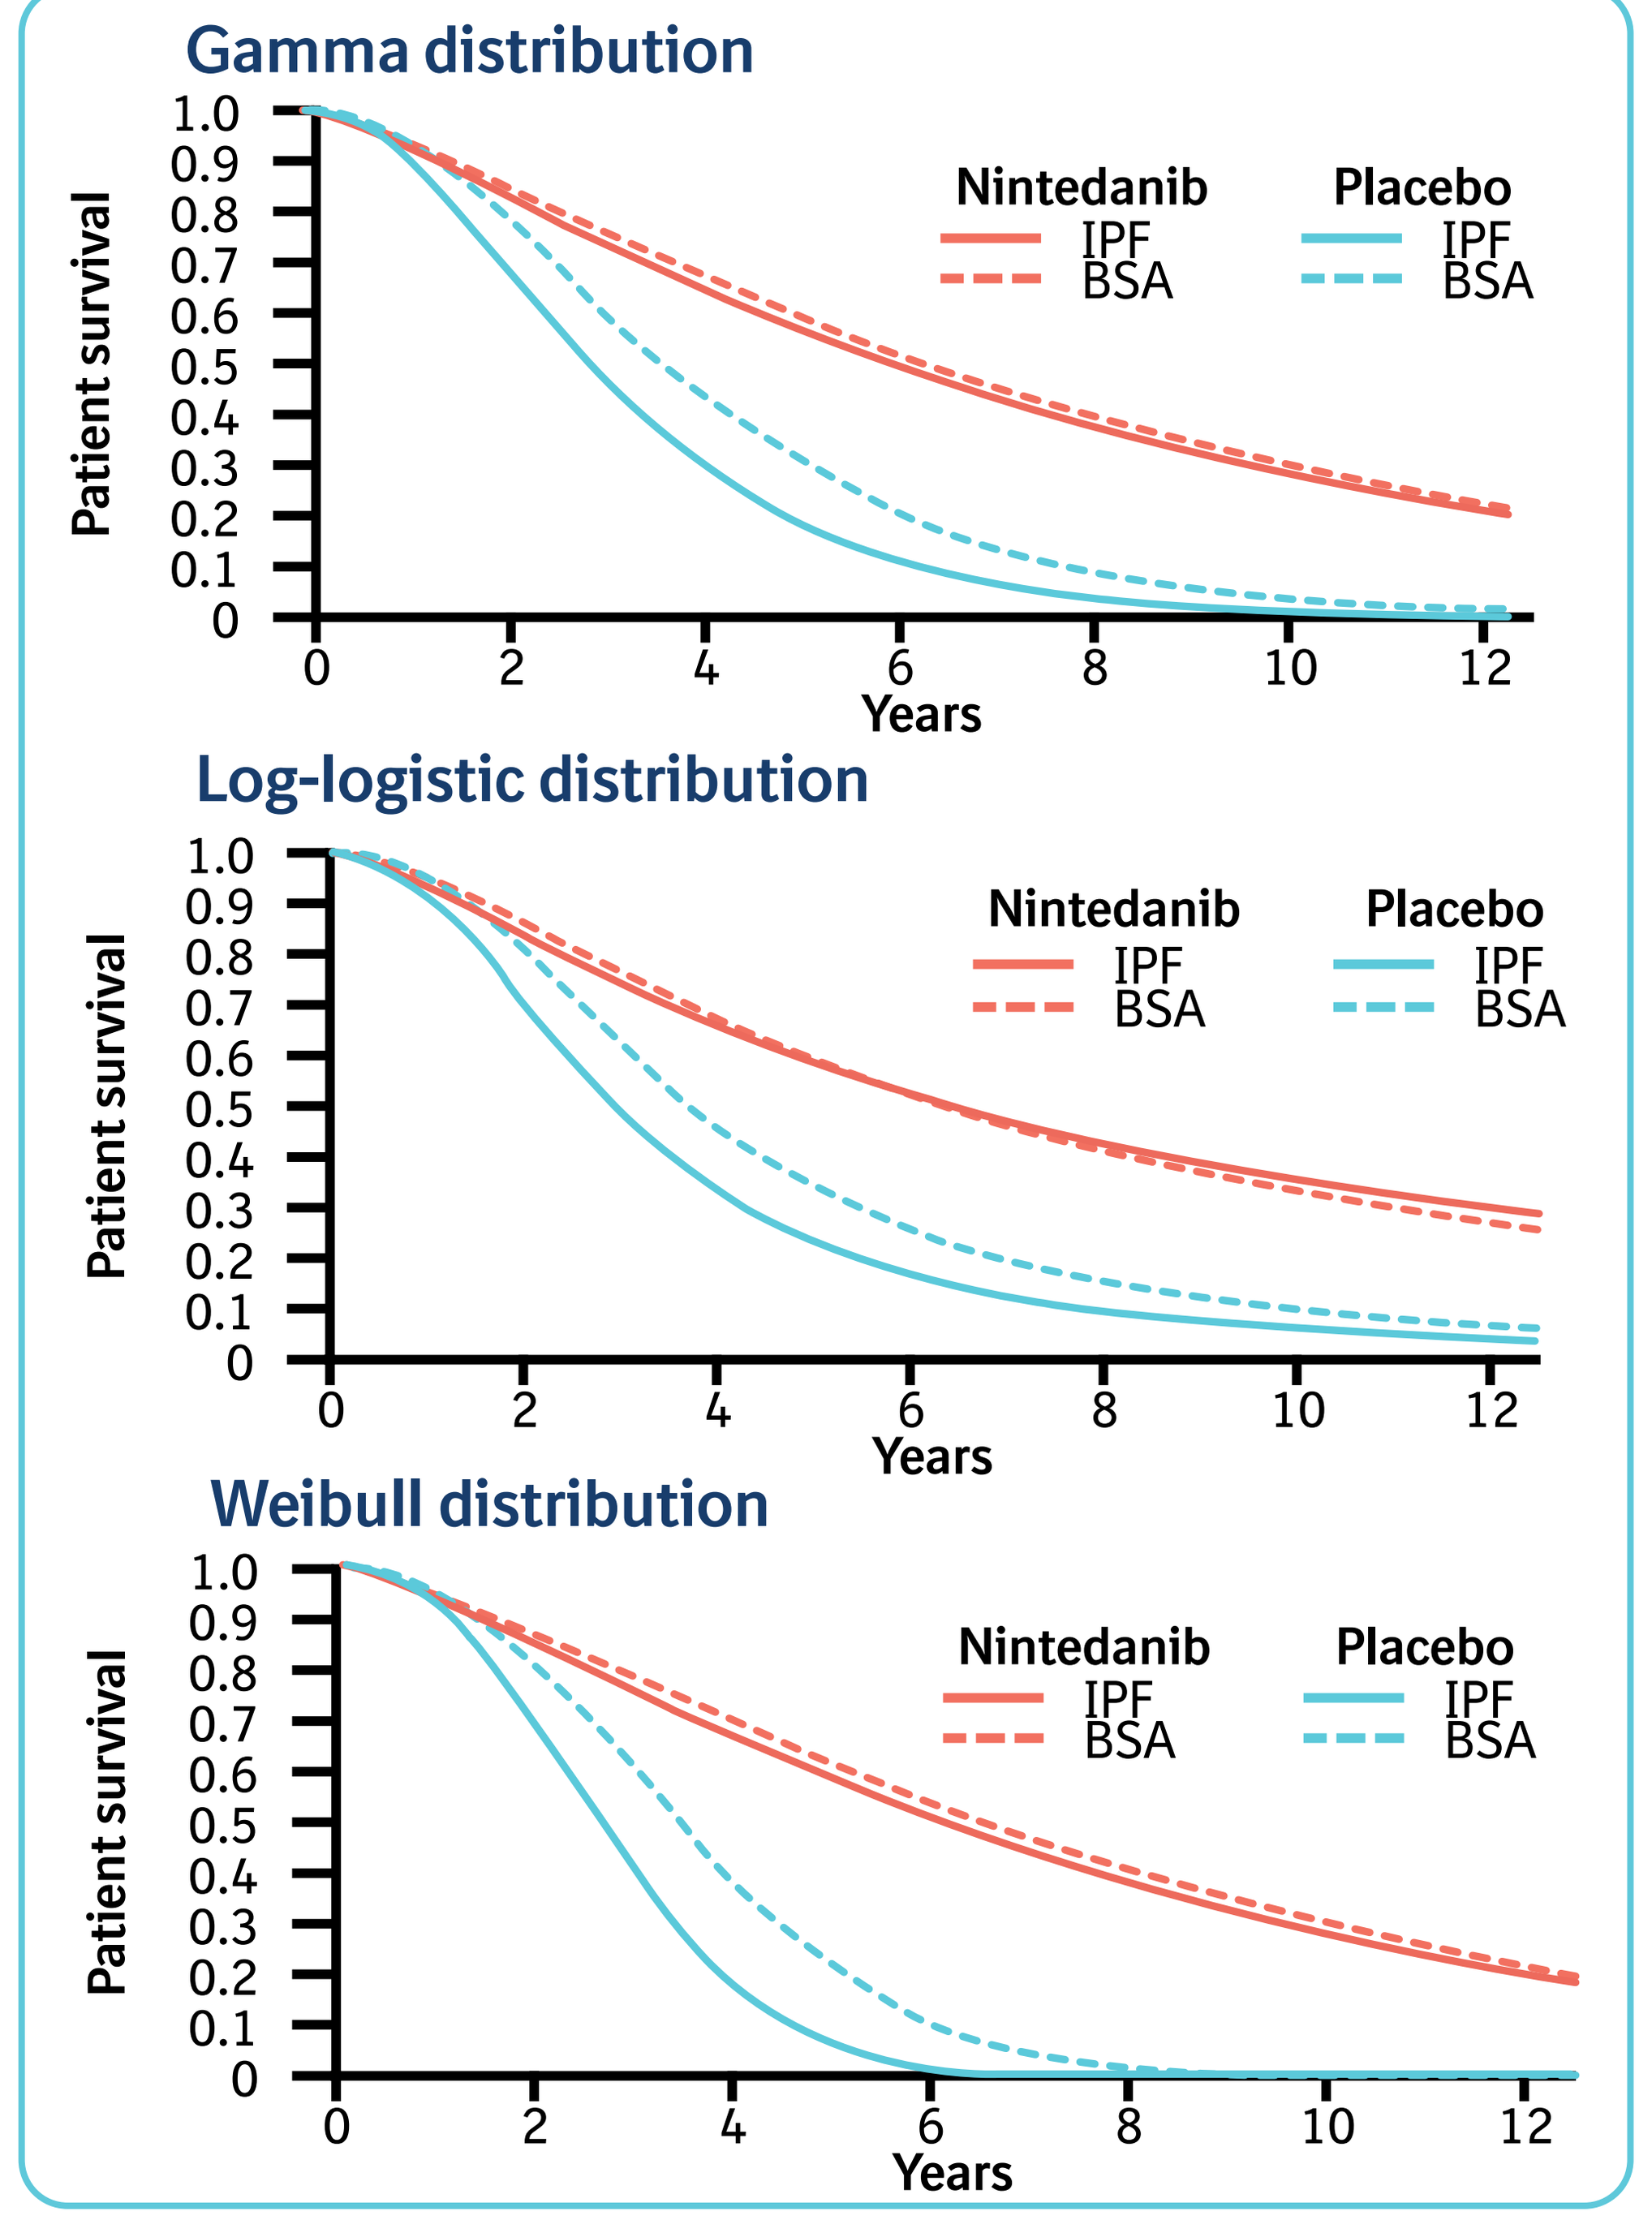
Gamma distribution
Log-logistic distribution
Weibull distribution
Nintedanib
IPF
BSA
Placebo
IPF
BSA
Patient survival
Years

Scan QR code or visit URL for a device-friendly version of this poster
https://bit.ly/3fe4als

INTERACTIVE

Scan QR code or visit URL for a webpage featuring all BI-supported publications at ERS 2021.
https://bit.ly/3BJKhwc

References

1. Flaherty KR, et al. N Engl J Med. 2019; 381:1718–1727;
2. Brown KK, et al. Eur Respir J. 2020; 55:2000085;
3. Simpson T, et al. Eur Respir J. 2021; 58:2100221;
4. Richeldi L, et al. Thorax. 2018; 73:581–583;
5. Richeldi L, et al. N Engl J Med. 2014; 370:2071–2082;
6. Crestani B, et al. Lancet Respir Med. 2019;7:60–68;
7. Behr J, et al. Eur Respir J 2020; 56:1902279;
8. Antoniou K, et al. ERJ Open Res 2020; 6:00172-2019;
9. Lancaster L, et al. BMJ Open Respir Res 2019; 6:e000397.

Abbreviations

BSA, Bayesian survival analysis; DL_{CO}, diffusing capacity of the lung for carbon monoxide; FVC, forced vital capacity; ILD, interstitial lung disease; IPF, idiopathic pulmonary fibrosis; OS, overall survival; PF-ILD, progressive fibrosing ILD.

Disclosures

BL and AD are employees of Symmetron Limited. TMM has received industry-academic funding from GlaxoSmithKline and UCB via his institution, and has received consultancy or speaker fees from Apellis, AstraZeneca, Bayer, Blade Therapeutics, Boehringer Ingelheim, Bristol-Myers Squibb, Galapagos, GlaxoSmithKline, Indalo, Novartis, Pliant, ProMetic, Respivant, Roche, Samumed and UCB outside the submitted work. YI has worked as an advisor, steering committee member and speaker for Asahi Kasei, Boehringer Ingelheim, Galapagos, NITTO, Promedior, Roche, Shionogi Savara and Taiho. KBR and MB are employees of Boehringer Ingelheim International GmbH.

Acknowledgements

The analysis was supported by Boehringer Ingelheim International GmbH (BI). The authors meet criteria for authorship as recommended by the International Committee of Medical Journal Editors (ICMJE). The authors did not receive payment for the development of the poster. Writing assistance was provided by Darren Chow, MSc, and Hannah Cook, PhD, of MediTech Media, and was contracted and funded by BI. BI were given the opportunity to review the poster for medical and scientific accuracy as well as intellectual property considerations.

Boehringer Ingelheim

ePoster to accompany an oral presentation at virtual ERS 2021, 5–8 September